CAPTIVATE Trial Design: A Pivotal Phase 3 Study of Claseprubart in Chronic Inflammatory Demyelinating Polyneuropathy

Jeffrey Allen,[1] Luis Querol,[2] Filip Eftimov,[3] Stojan Peric,[4] Tina Dysgaard,[5] Yusuf A. Rajabally,[6] Thomas Harbo,[7] Eduardo Nobile-Orazio,[8] Hans Katzberg,[9] Jon Katz,[10] James K. Sheffield,[11] Luke Hickey,[11] Nuria Carrillo,[11] Richard Lewis[12]

[1]University of Minnesota, Minneapolis, MN, USA; [2]Hospital de la Santa Creu, Barcelona, Spain; [3]Amsterdam UMC, Amsterdam, The Netherlands; [4]University of Belgrade, Belgrade, Serbia; [5]University of Copenhagen, Copenhagen, Denmark; [6]University of Aston, Birmingham, UK; [7]Aarhus University Hospital, Aarhus, Denmark; [8]Università degli Studi di Milano, Milan, Italy; [9]Institute of Medical Science, Toronto, Canada; [10]California Pacific Medical Center, San Francisco, CA, USA; [11]Dianthus Therapeutics, New York, NY, USA; [12]Cedars Sinai, Los Angeles, CA, USA.

004

scimobi

MAIN FINDINGS

- CAPTIVATE is an innovative, rigorous phase 3 study evaluating claseprubart, an active C1s inhibitor administered as a convenient subcutaneous biweekly injection, in CIDP.
- CAPTIVATE features a relapse-prevention design, commonly used in studies of interventions in CIDP.
- CAPTIVATE prioritizes patient safety, avoids complete washout of standard of care, minimizes exposure to ineffective therapy, and provides continued access as part of an optional open-label extension.

INTRODUCTION

- Chronic inflammatory demyelinating polyneuropathy (CIDP) is a chronic, progressive, acquired autoimmune disease targeting peripheral nerves, resulting in demyelination.
- Current standard of care for CIDP includes intravenous (IV) or subcutaneous (SC) immunoglobulins (Ig), corticosteroids, and plasmapheresis, all of which are associated with limited long-term efficacy or burdensome administration.[1]
- Several complement inhibitors are in development for the treatment of CIDP and increasing evidence suggests that activation of the classical complement pathway (CCP) plays a crucial role in the pathophysiology of CIDP.[2]
- Claseprubart (DNTH103) is an investigational, clinical-stage, potent monoclonal antibody engineered to selectively target the CCP by inhibiting the active form of the C1s protein, a clinically validated complement target (**Figure 1**). Claseprubart is designed to enable a more convenient SC, infrequently dosed, self-administered injection.
- In the phase 2 MaGic Study, SC Q2W claseprubart demonstrated clinically meaningful improvements in patients with generalized myasthenia gravis with a well-tolerated safety profile,[3] supporting the therapeutic potential of active C1s blockade in complement-mediated autoimmune neurological disorders such as CIDP.
- Here, we describe the design of the ongoing CAPTIVATE study, evaluating the efficacy and safety of claseprubart in patients with CIDP.

Figure 1. Claseprubart Mechanism of Action.

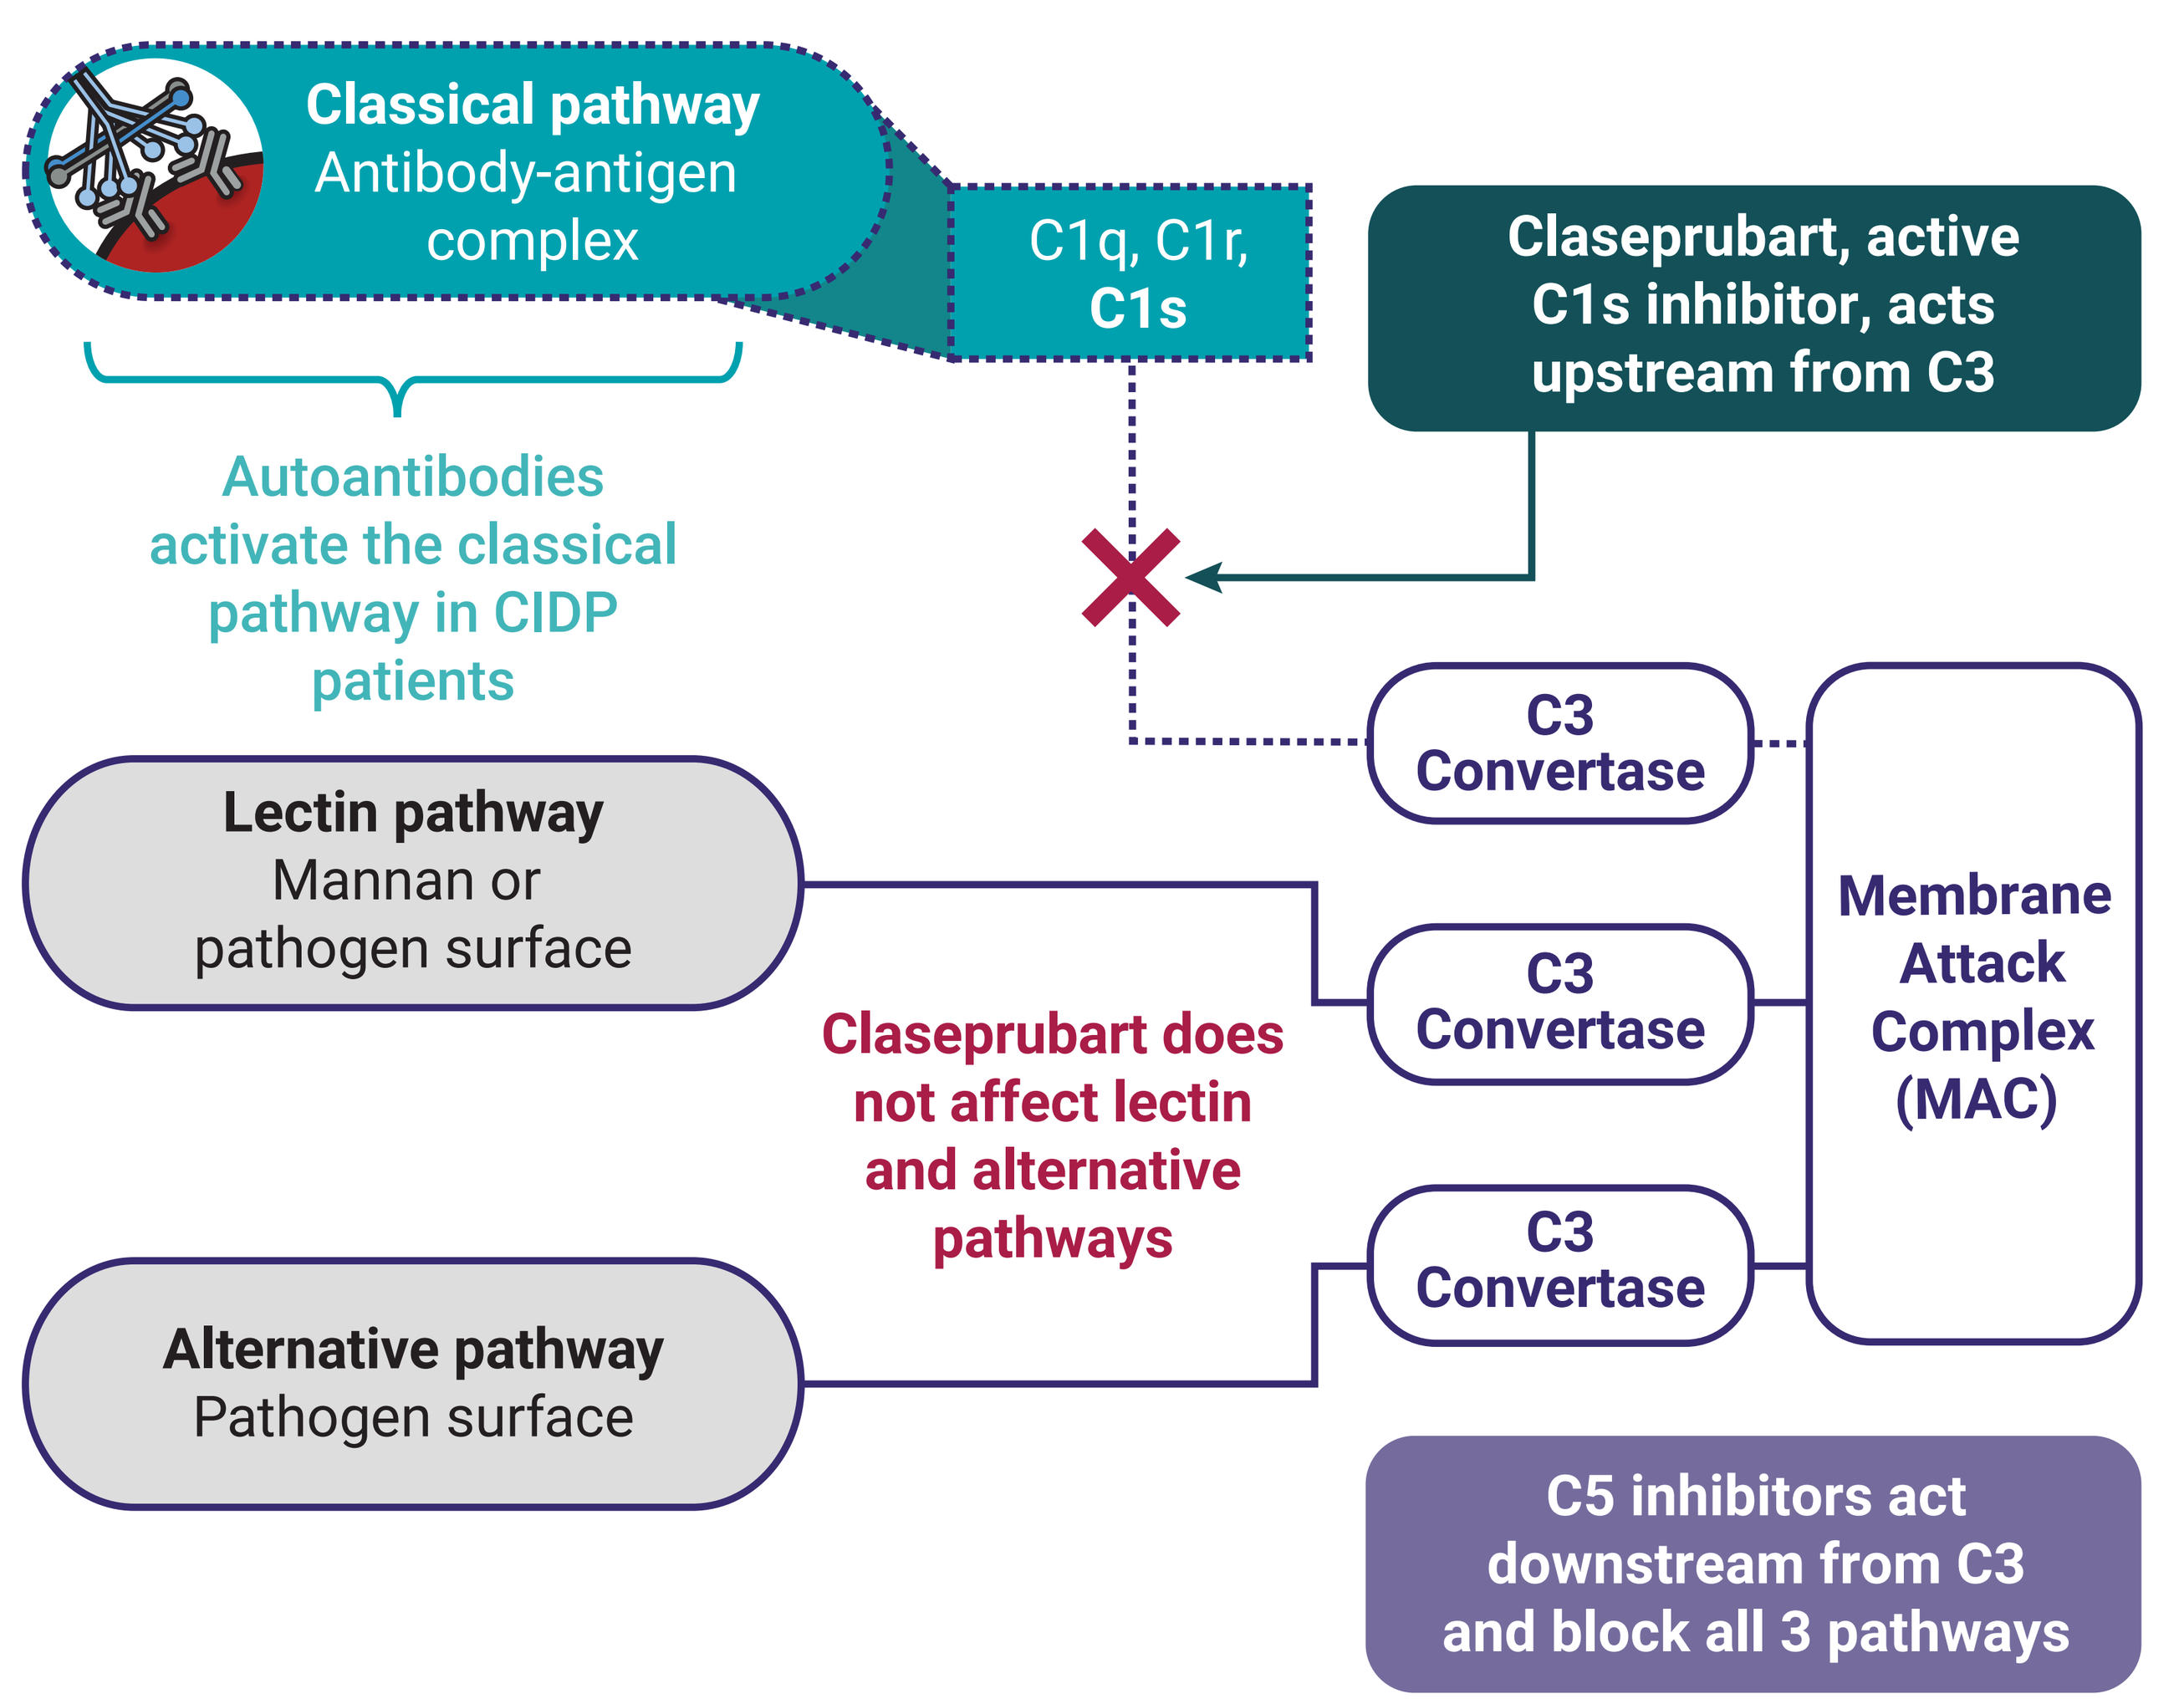
Classical pathway
Antibody-antigen complex
C1q, C1r, C1s
Claseprubart, active C1s inhibitor, acts upstream from C3
Autoantibodies activate the classical pathway in CIDP patients
Lectin pathway
Mannan or pathogen surface
Claseprubart does not affect lectin and alternative pathways
C3 Convertase
C3 Convertase
C3 Convertase
Membrane Attack Complex (MAC)
Alternative pathway
Pathogen surface
C5 inhibitors act downstream from C3 and block all 3 pathways

CIDP, chronic inflammatory demyelinating polyneuropathy

Figure 2. CAPTIVATE Study Design.

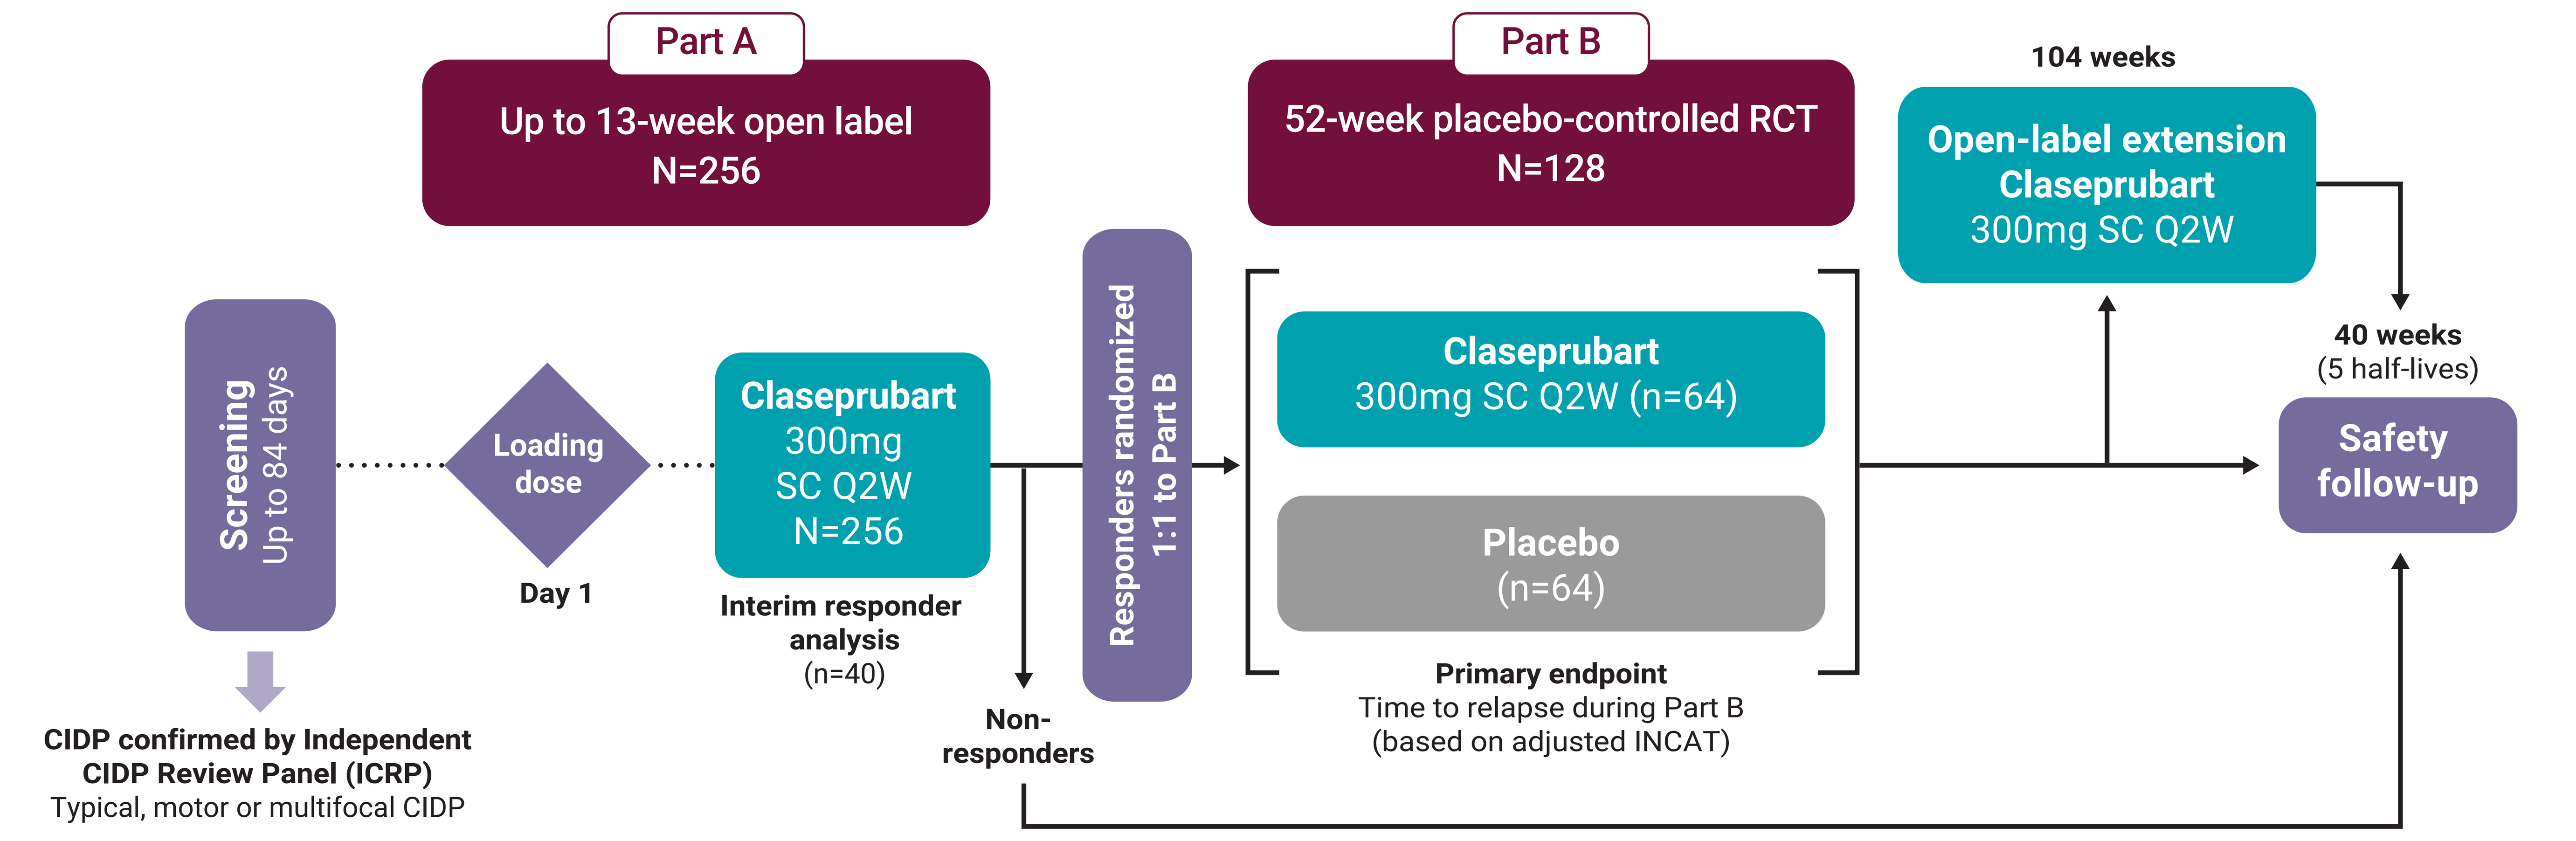
Part A
Up to 13-week open label
N=256
Part B
52-week placebo-controlled RCT
N=128
104 weeks
Open-label extension
Claseprubart
300mg SC Q2W
Screening
Up to 84 days
Loading dose
Day 1
Claseprubart
300mg
SC Q2W
N=256
Interim responder analysis
(n=40)
Responders randomized 1:1 to Part B
Claseprubart
300mg SC Q2W (n=64)
Placebo
(n=64)
40 weeks
(5 half-lives)
Safety follow-up
CIDP confirmed by Independent CIDP Review Panel (ICRP)
Typical, motor or multifocal CIDP
Non-responders
Primary endpoint
Time to relapse during Part B
(based on adjusted INCAT)

Patients aged 18–75 years with confirmed CIDP are administered and IV loading dose of claseprubart followed by 300mg SC Q2W in Part A. Patients who responded (≥1 point decrease in INCAT score) were eligible for Part B and randomized to receive 300mg claseprubart SC Q2W or placebo for 52 weeks. CIDP, chronic inflammatory demyelinating polyneuropathy; INCAT, Inflammatory Neuropathy Cause and Treatment; IV, intravenous; Q2W, once every 2 weeks; RCT, randomized controlled trial; SC, subcutaneous

STUDY DESIGN AND DOSING

- CAPTIVATE is a global, multicenter, randomized, double-blind, placebo-controlled phase 3 study (NCT06858579) that is evaluating claseprubart in a broad population of adults with a diagnosis of CIDP.

Part A Design

- Part A is a 13-week open-label period in which patients are administered SC 300mg claseprubart Q2W for 13 weeks following an IV loading dose (**Figure 2**).
 - Beginning at Week 5, patients will be assessed for a response, defined as a ≥1 point decrease in the adjusted INCAT (Inflammatory Neuropathy Cause and Treatment) score compared to Part A baseline. If the response is confirmed at the next consecutive visit, the participant will enter Part B (if the patient continues to meet the definition for response at Day 1 Part B). Non-responders will enter the safety follow-up.
 - It is anticipated that approximately 256 participants will be enrolled into Part A. An interim responder analysis was planned with the first 40 patients completing Part A.

Part B Design

- Part B is a randomized, placebo-controlled, double-blind treatment period of up to 52 weeks for participants who respond to claseprubart in Part A.
 - Approximately 128 participants identified as responders in Part A will be randomized 1:1 in Part B to receive 300mg claseprubart or placebo given SC once every 2 weeks.

Open-Label Extension and Safety Follow-up

- Patients who complete Part B are permitted to enter a 104-week open-label extension (OLE). Additionally, patients who have a relapse in Part B (increase of ≥1 point in adjusted INCAT score) are also eligible for the OLE, including those who received Ig, corticosteroid, or PLEX rescue therapy.
- All patients are eligible for a 40-week safety follow-up period following their last dose of claseprubart or placebo. Patients can resume standard of care treatment for CIDP per the investigator's judgment.

Study Population

- Adults aged 18–75 years (inclusive) with a diagnosis of CIDP or possible CIDP per the 2021 EAN/PNS guidelines.[4] Participants must have either typical, motor or multifocal CIDP.
- At screening, patients must have a CIDP Disease Activity Status (CDAS) score ≥3, and INCAT score between 2 and 9 (inclusive) and be neurologically stable.
- Patients on prior standard of care (SoC) therapy were eligible if they are SoC-refractory (worsening or inadequate response to SoC) or SoC-treated (current or past treatment with SoC). If applicable, oral corticosteroids (after tapering) and allowed immunosuppressants are continued in Parts A and B. Treatment-naive patients are also eligible.
- For patients on IV or SC Ig, the last dose for both must occur 1 week prior to Day 1 of Part A. Patients who received complement inhibitors or anti-CD20 treatments within the previous 6 months are not eligible.

Assessments

- The primary objective of CAPTIVATE is to evaluate the efficacy of claseprubart compared to placebo, assessed as the time from first dose in Part B to relapse (≥1 point increase in adjusted INCAT) (**Table 1**).

Table 1. Assessments and Endpoints

Efficacy assessments	
Secondary	• Time from first dose in Part B to a decrease from Part B baseline of ≥4 points (centile metric) in I-RODS score during Part B • Time from first dose in Part B to a decrease from Part B baseline of ≥8 kPa in grip strength in the dominant hand during Part B • Health-related quality of life outcomes
Primary	• Evaluate efficacy of claseprubart compared to placebo, assessed as the time from first dose in Part B to relapse (≥1 point increase in adjusted INCAT)
Other assessments	
• Safety and tolerability, including adverse events • Pharmacokinetics and pharmacodynamics • Immunogenicity of claseprubart • Assess effect of claseprubart on biomarkers of disease activity	

INCAT, Inflammatory Neuropathy Cause and Treatment; I-RODS, Rasch-built Overall Disability Scale

- Safety and tolerability will be assessed throughout the study and OLE.

INTERIM RESPONDER ANALYSIS

- CAPTIVATE was launched in 2024 and is actively enrolling participants in North America, South America, Europe, and Asia.
 - In March 2026, a planned interim responder analysis was conducted on currently enrolled patients in Part A.[5] The target was a response rate of 50% or greater (i.e., ≥20 confirmed responders out of first 40 participants to complete Part A) based on precedent set with aC1s inhibition.
 - The decision to continue to Part B of the study was reached early, after 20 confirmed responders were achieved with less than 40 planned participants completing open-label Part A of the trial.

References

1. Ryan M, Ryan SJ. Am J Manag Care 2018;24:S371–S9
2. Querol LA, et al. Neurotherapeutics 2022;19:864–73
3. Narayanaswami P, et al. AANEM. San Francisco, CA. 2025.
4. Van den Bergh PYK, et al. Eur J Neurol 2021;28:3556–83
5. Dianthus Therapeutics Inc. 2026. https://investor.dianthustx.com

Acknowledgements

The authors thank Aji Nair of Dianthus Therapeutics Inc. for assistance with the preparation of this poster, and Eastmond Medicomm Ltd for editing and production support, funded by Dianthus Therapeutics Inc.

Disclosures

All authors except **JKS, LH, BB** and **NC** have received grants and/or honoraria from Dianthus Therapeutics Inc. **JKS, LH, BB** and **NC** are employees of Dianthus.

Presented at the AAN Autoimmune Neurology Conference (ANC) , 7–8 August, 2026, Houston, TX, USA

Dianthus Therapeutics Inc., New York, NY, USA