Potential Mechanistic Advantages of aC1s Targeting in Myasthenia Gravis: Upstream Versus Downstream Complement Blockade

058

Tuan Vu,[1] Marianna Lalla,[2] Yang Zhao,[2] Linda Rehaume,[2] Jennifer Cross,[2] Shahar Shelly[3]

[1]University of South Florida, FL, USA; [2]Dianthus Therapeutics, NY, USA; [3]Department of Neurology, Rambam Health Care Campus, Haifa, Israel.

MAIN FINDINGS

- Both ravulizumab and claseprubart inhibited MAC formation as the terminal event in the classical pathway.
- However, only upstream aC1s inhibition with claseprubart and not ravulizumab inhibited C3a formation and C3b deposition.
- These findings suggest that upstream inhibition with claseprubart at the level of aC1s has greater potential for broad inflammatory control in gMG than C5 blockade and supporting the possibility for added clinical benefit while sparing the alternative and lectin pathways.

INTRODUCTION

- In generalized myasthenia gravis (gMG), pathogenic antibodies trigger classical complement pathway (CCP) activation, resulting in membrane attack complex (MAC)-mediated neuromuscular junction (NMJ) injury and progressive disability.[1,2]
- Claseprubart (DNTH103) is an investigational monoclonal antibody that specifically blocks the CCP by selectively binding active C1s (aC1s).
 - Unlike currently approved complement inhibitors such as C5 inhibitor ravulizumab, claseprubart offers a more focused treatment approach in gMG by only blocking the CCP while preserving the lectin and alternative pathway (**Figure 1**).
 - The lectin and alternative complement pathways play a significant role in anti-bacterial immunity and complement inhibitor therapies that maintain their function may reduce the risk of severe encapsulated bacterial infections.[3]

Figure 1. Claseprubart Mechanism of Action.

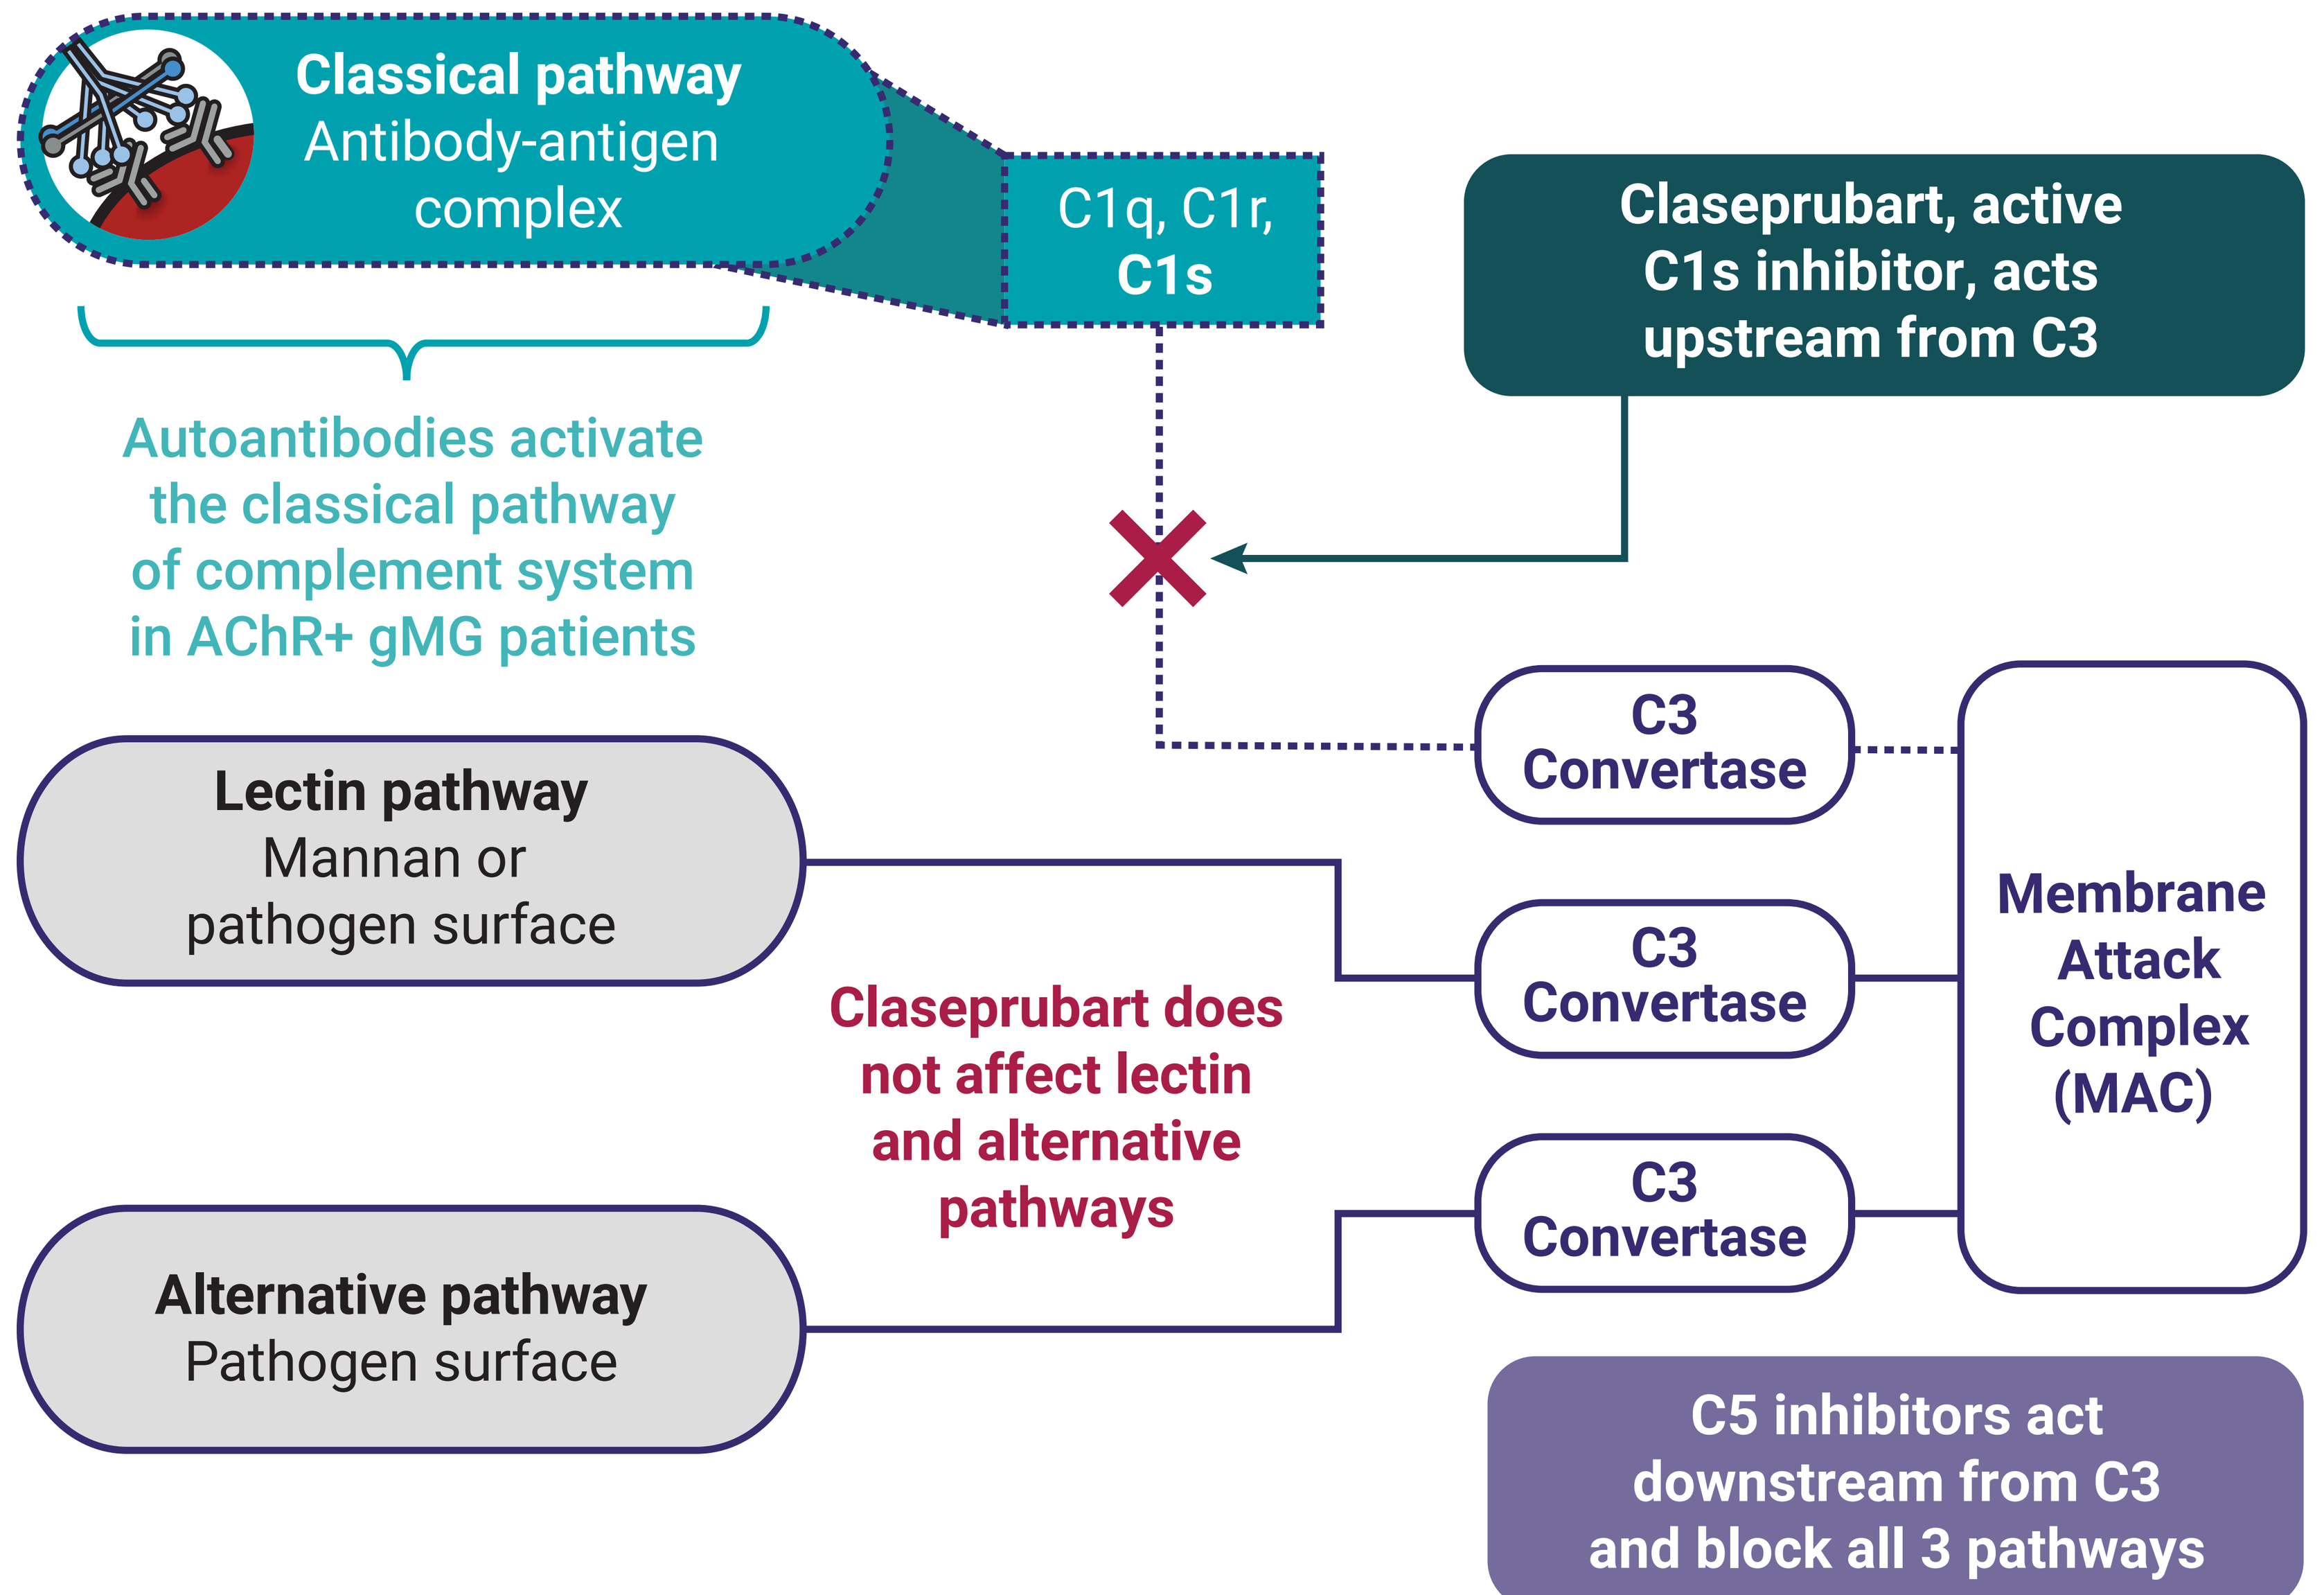
Classical pathway
Antibody-antigen complex
C1q, C1r, C1s
Claseprubart, active C1s inhibitor, acts upstream from C3
Autoantibodies activate the classical pathway of complement system in AChR+ gMG patients
Lectin pathway
Mannan or pathogen surface
Claseprubart does not affect lectin and alternative pathways
Alternative pathway
Pathogen surface
C3 Convertase
C3 Convertase
C3 Convertase
Membrane Attack Complex (MAC)
C5 inhibitors act downstream from C3 and block all 3 pathways

AChR+ gMG, acetylcholine receptor-positive generalized myasthenia gravis.

OBJECTIVE

- To determine whether aC1s inhibition by claseprubart suppresses generation of C3a, C3b and membrane attack complex (MAC) *in vitro*, and whether its complement inhibition profile is different to that of the C5 inhibitor ravulizumab.

METHODS

Three assays were used to characterize the inhibition of aC1s, C3a and C3b by claseprubart and ravulizumab.

WIESLAB® Classical Complement Assay (Svar Life Science, Malmö Sweden)

- Assesses functional activity of CCP in human serum.
- ELISA (enzyme-linked immunosorbent assay)-based functional assay using IgM-coated microtiter wells to trigger classical pathway activation in diluted human serum, leading to formation of the complement components, including MAC.
- MAC formation quantified by anti-C5b-9 alkaline phosphatase-conjugated antibody and p-nitrophenyl phosphate substrate, with absorbance measured at 405nm.

Human C3a ELISA (Hycult Biotech, East Hartford, USA)

- Sandwich ELISA employing capture antibodies and biotinylated detection antibodies specific for a C3a-desArg neoepitope, enabling selective measurement of C3a without cross-reactivity with intact C3.
- Colorimetric detection using streptavidin-peroxidase and tetramethylbenzidine substrate, with absorbance read at 450nm and concentrations determined from a C3a standard curve.

C3b Red Blood Cell Deposition Assay (Abcam, Waltham, USA)

- Cell-based functional assay using antibody-sensitized human red blood cells (RBCs) to activate the classical complement pathway and induce surface deposition of C3b.
- Flow cytometric quantification of C3b deposition using fluorescein isothiocyanate-labelled anti-C3c antibody, with premature termination of the cascade to prevent hemolysis and preserve intact cells.
- For each assay, we tested dilution series of claseprubart, ravulizumab, and an isotype control. Each experiment was repeated three times.

RESULTS

- In head-to-head WIESLAB® Complement Classical Pathway assays, claseprubart produced comparable inhibition of MAC formation compared to ravulizumab, confirming similar levels of terminal pathway blockade (**Figure 2**).
- In contrast, only upstream aC1s inhibition with claseprubart resulted in near complete inhibition of the generation of C3a, while ravulizumab did not (**Figure 3**).
- Similarly, in the C3b assay, C3b-positive RBCs were all but undetectable in claseprubart-treated assay samples, but levels were unaffected in samples treated with ravulizumab (**Figure 4**).

Figure 2. Classical Pathway Inhibition.

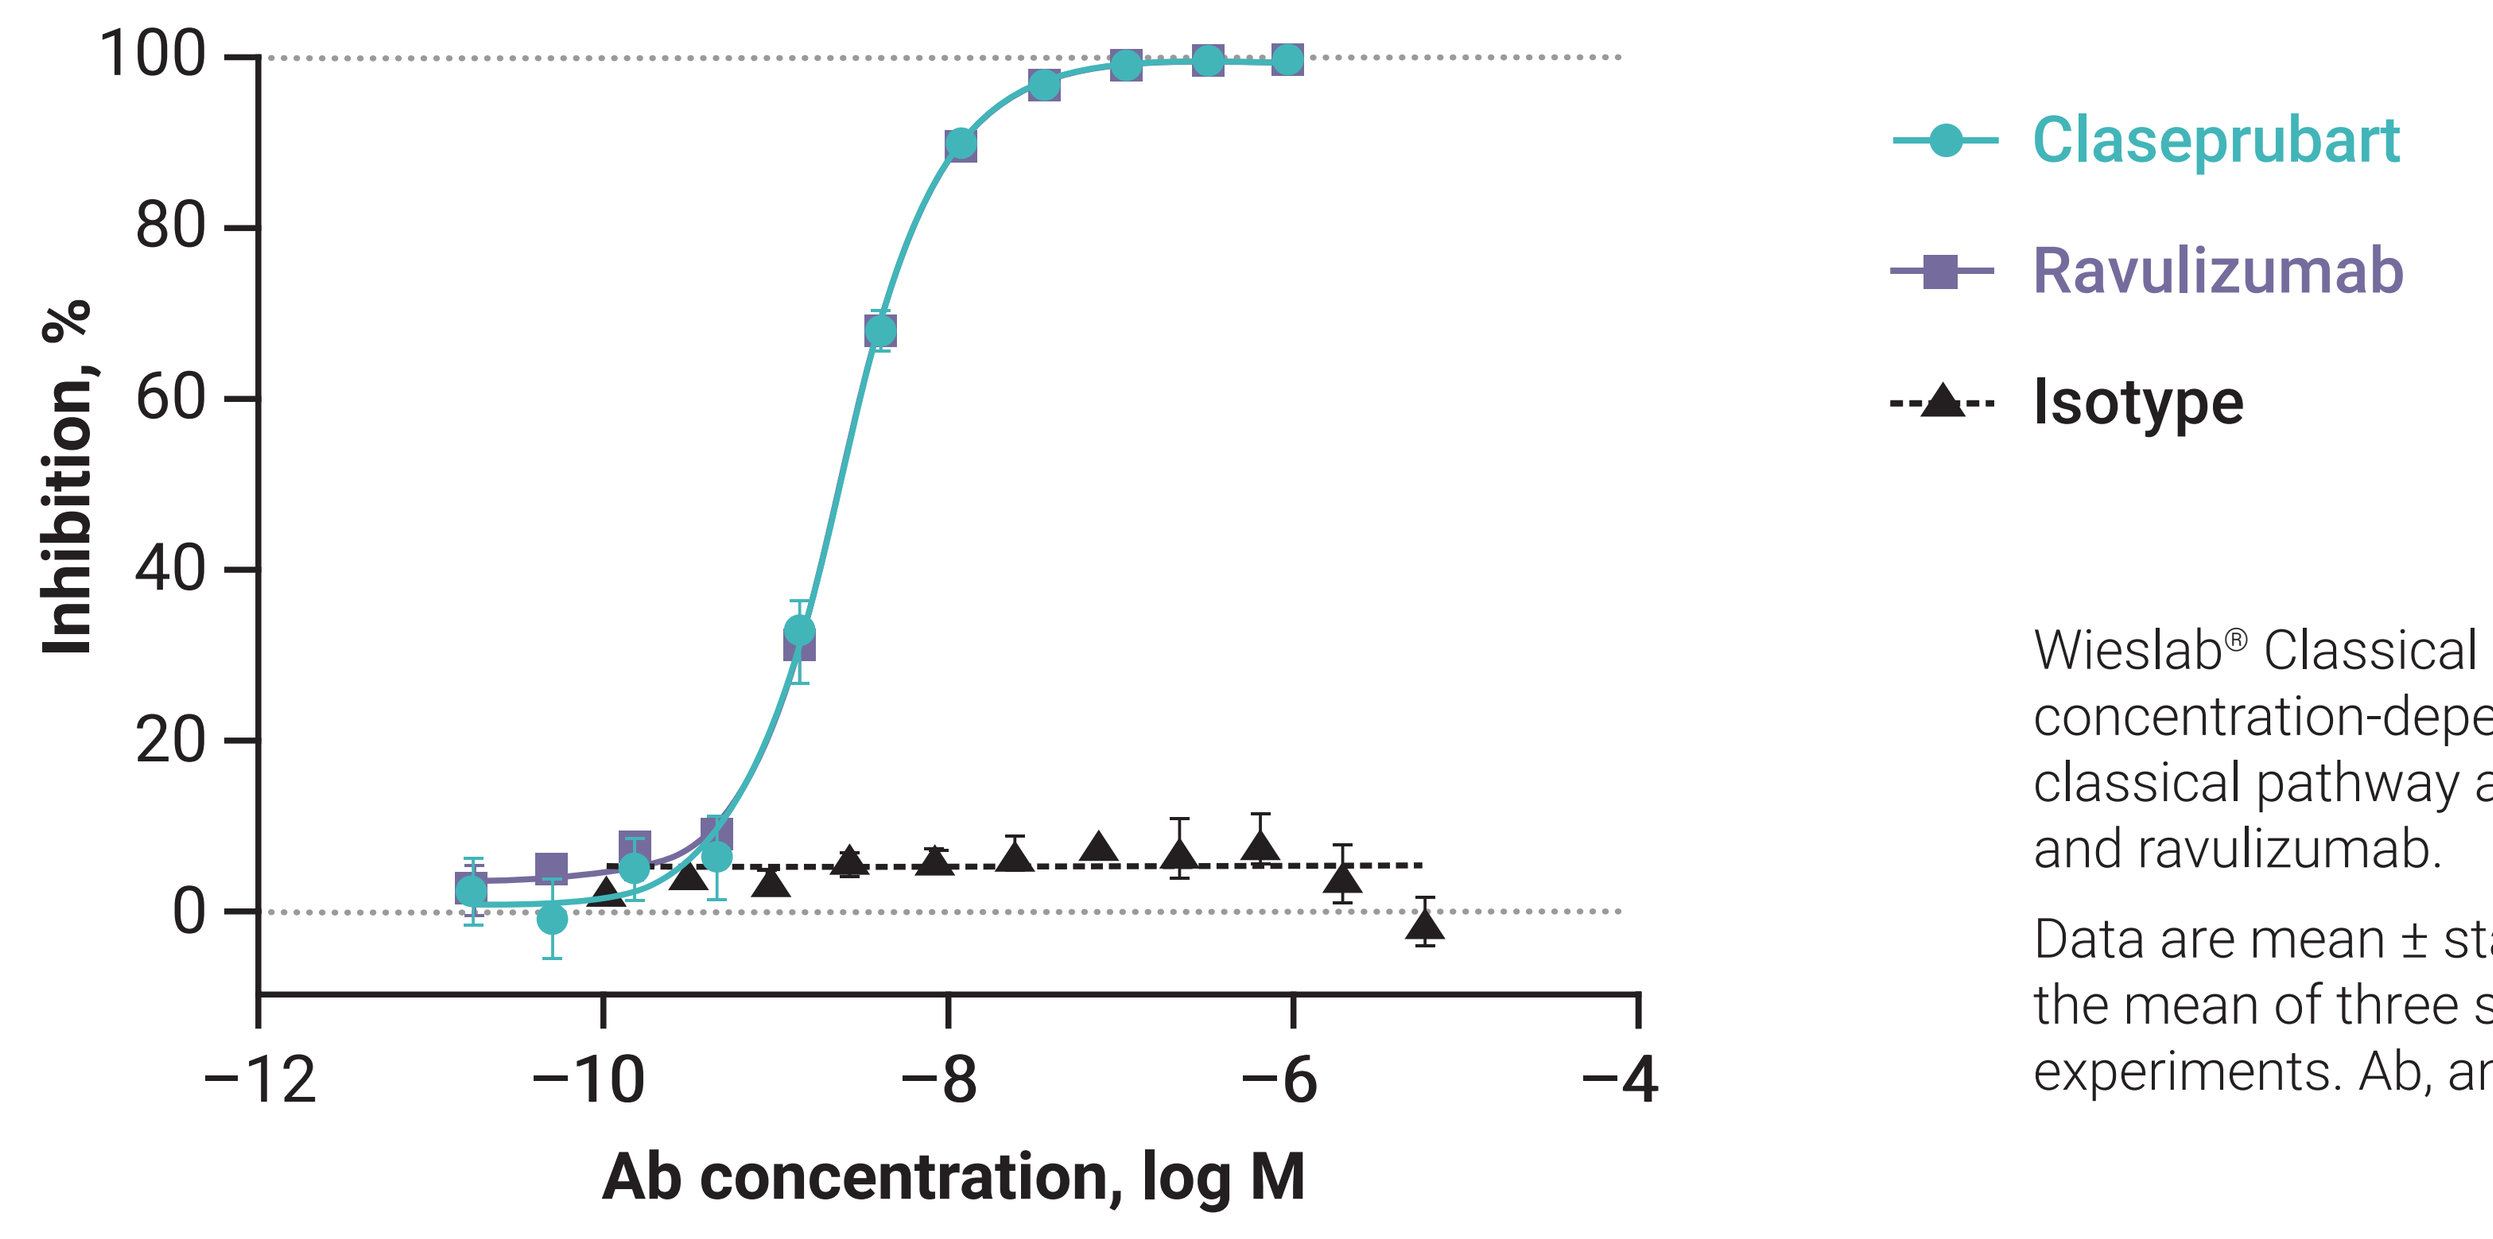
Inhibition, %
100
80
60
40
20
0
−12
−10
−8
−6
−4
Ab concentration, log M
Claseprubart
Ravulizumab
Isotype

Wieslab® Classical Pathway Assay showing concentration-dependent inhibition of classical pathway activity by claseprubart and ravulizumab.

Data are mean ± standard error of the mean of three separate *in vitro* experiments. Ab, antibody.

Figure 3. C3a Formation.

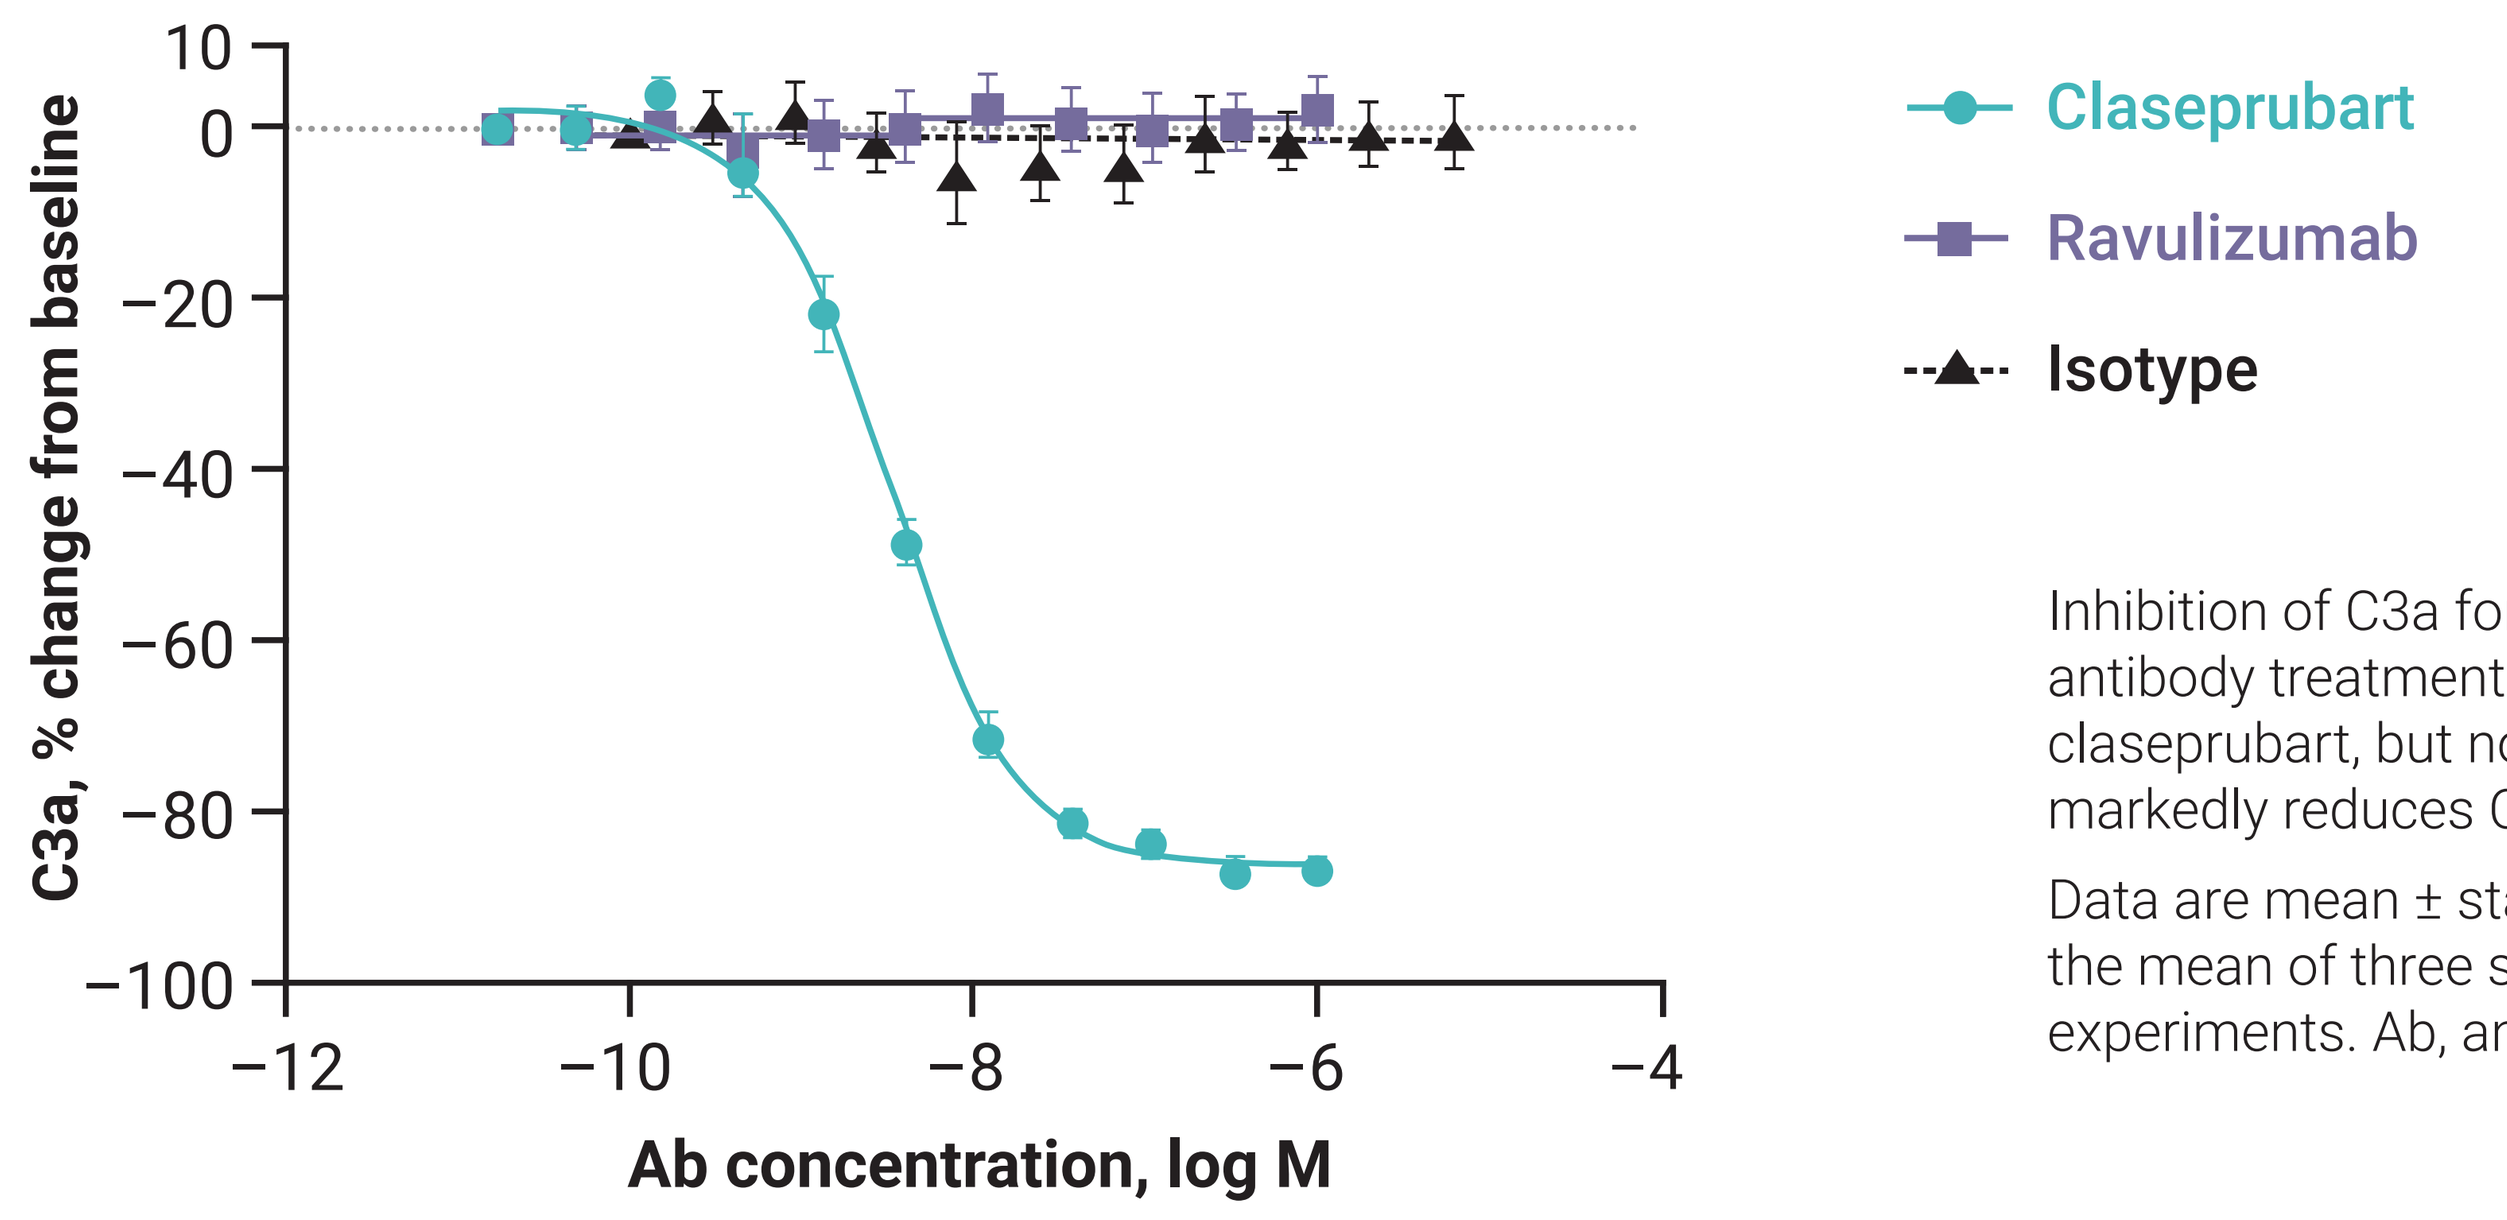
C3a, % change from baseline
10
0
−20
−40
−60
−80
−100
−12
−10
−8
−6
−4
Ab concentration, log M
Claseprubart
Ravulizumab
Isotype

Inhibition of C3a formation following antibody treatment demonstrates that claseprubart, but not ravulizumab, markedly reduces C3a production.

Data are mean ± standard error of the mean of three separate *in vitro* experiments. Ab, antibody.

Figure 4. C3b Formation.

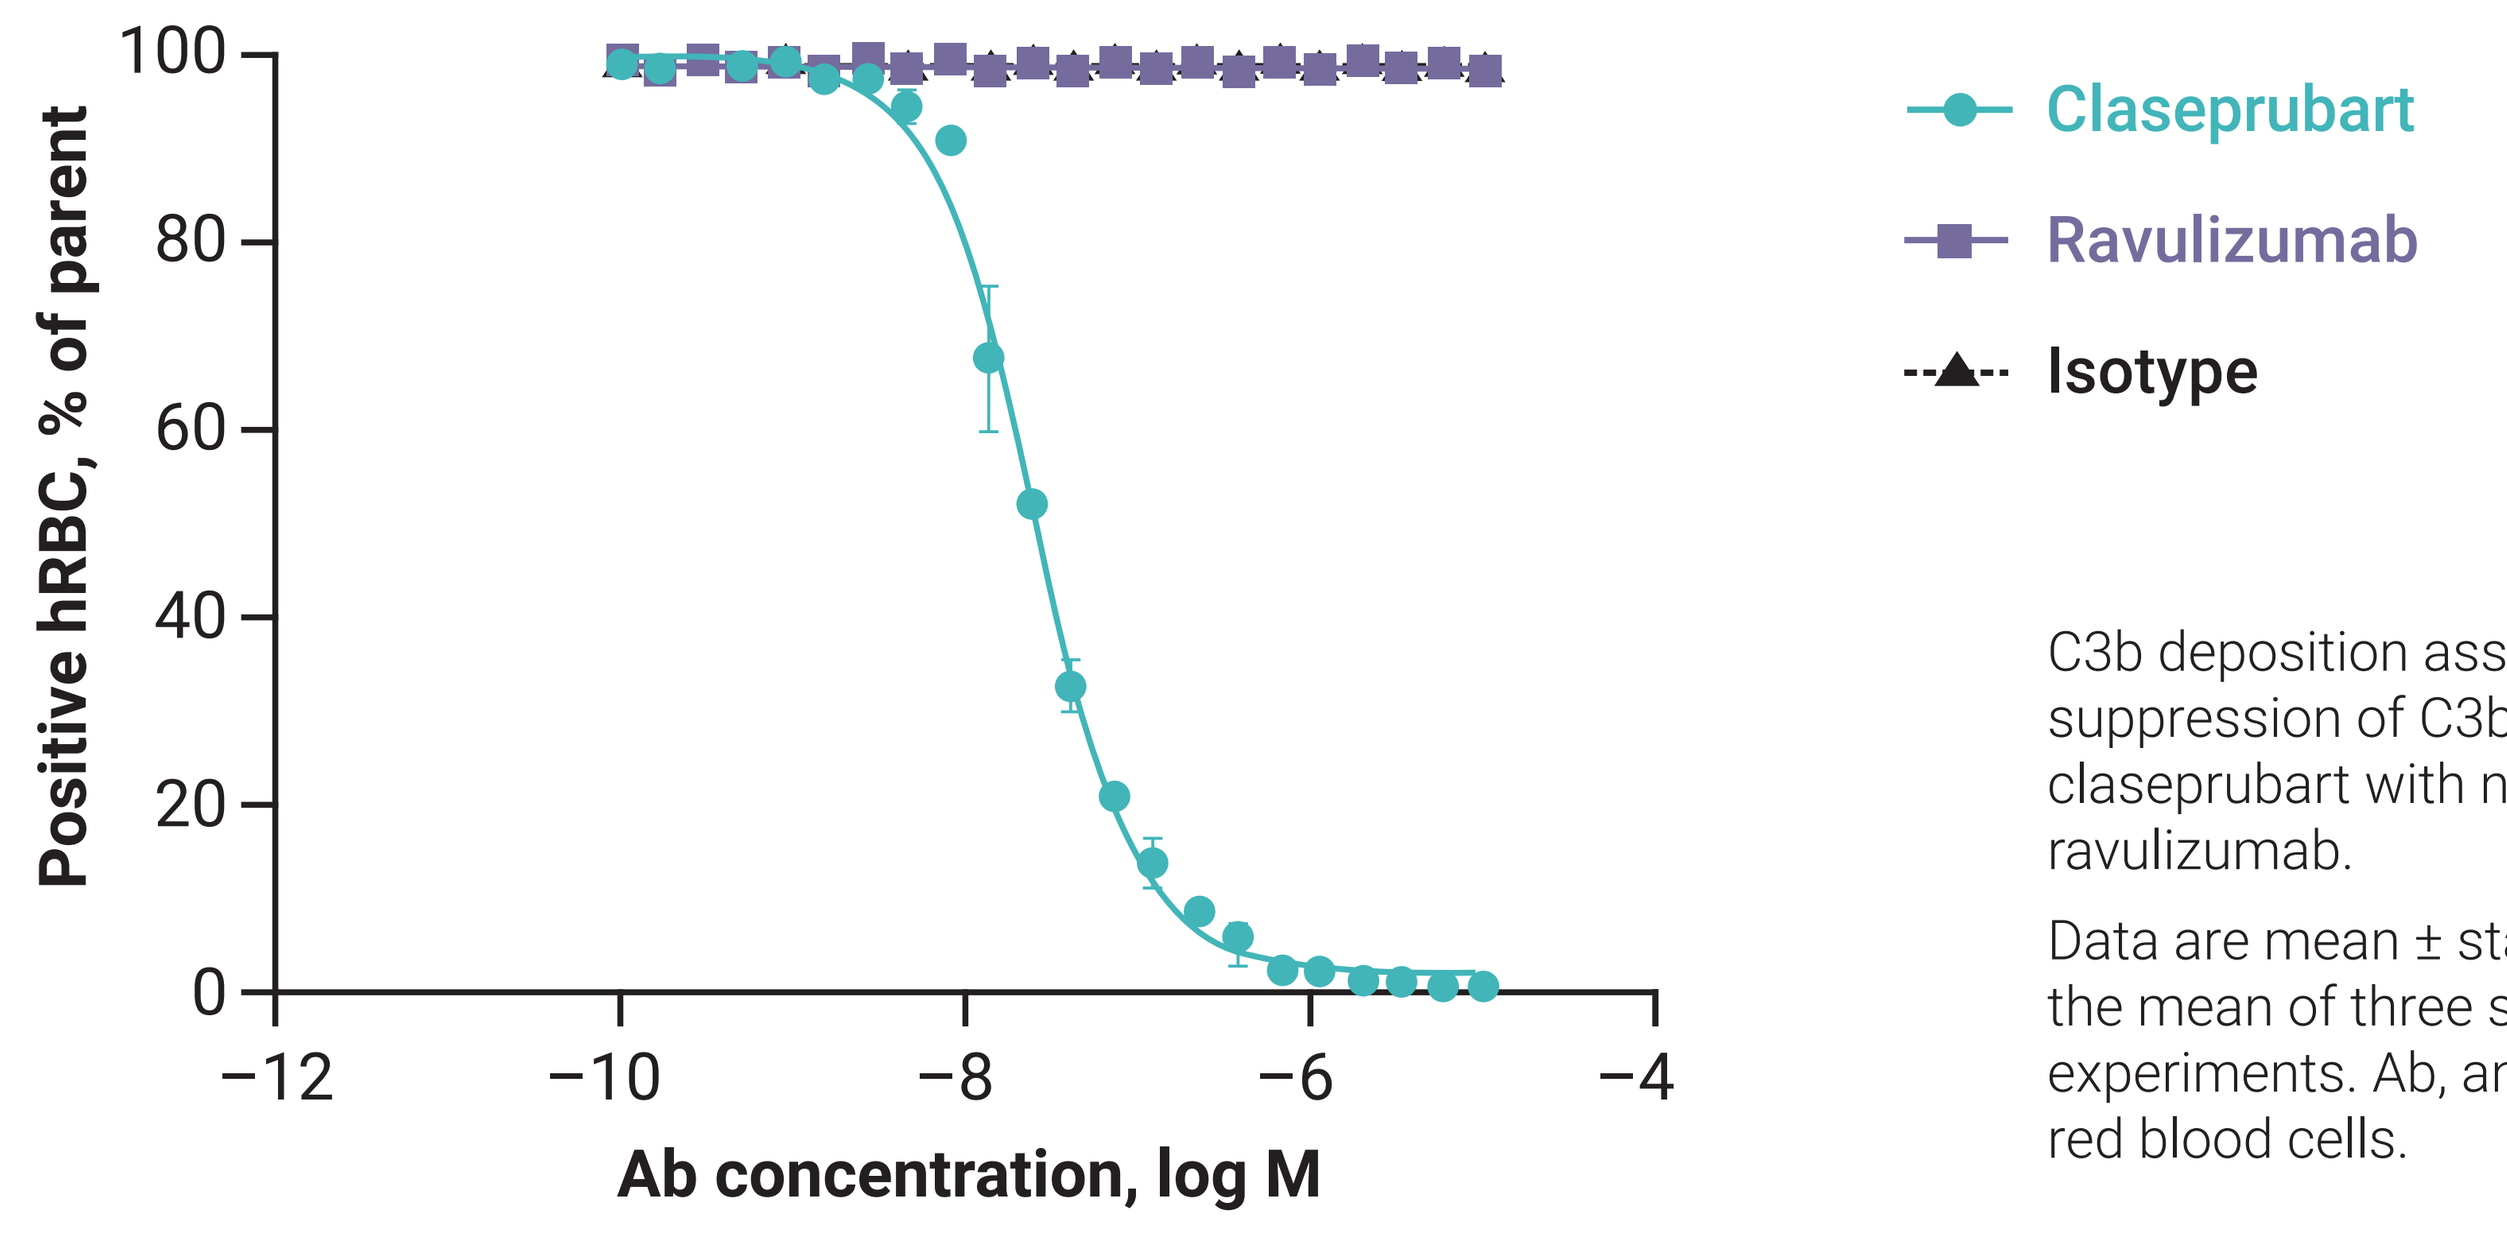
Positive hRBC, % of parent
100
80
60
40
20
0
−12
−10
−8
−6
−4
Ab concentration, log M
Claseprubart
Ravulizumab
Isotype

C3b deposition assay showing robust suppression of C3b generation by claseprubart with no effect observed for ravulizumab.

Data are mean ± standard error of the mean of three separate *in vitro* experiments. Ab, antibody; hRBC, human red blood cells.

References

1. Howard JF. Ann N Y Acad Sci 2018;1412:113–28
2. Sanderson NSR. Mol Immunol 2022;151:11–8
3. Halacova N, et al. Biologics 2026;6:3–21

Acknowledgements

The authors thank Aji Nair of Dianthus Therapeutics Inc. for assistance with the preparation of this poster, and Eastmond Medicomm Ltd for editing and production support, funded by Dianthus Therapeutics Inc.

Disclosures

SS and **TV** have received grants, honoraria and/or consultancy fees from Dianthus. **ML**, **YZ**, **LH** and **JC** are employees of Dianthus.

Presented at the AAN Autoimmune Neurology Conference (ANC) , 7–8 August, 2026, Houston, TX, USA

Dianthus Therapeutics Inc., New York, NY, USA