015

Safety and Efficacy of Claseprubart, an Active C1s Inhibitor, in Patients with Generalized Myasthenia Gravis

Tuan Vu,[1] Stojan Peric,[2] Pushpa Narayanaswami,[3] Marek Śmilowski,[4] Agnieszka Slowik,[5] Sankalp Gokhale,[6] Caitlin M. Briggs,[6] Uzma Siddiqui,[6] Simrat Randhawa,[6] Matt Truman,[7] Luke Hickey,[6] Shahar Shelly,[8] John Vissing[9]

[1]University of South Florida, Tampa, FL, USA; [2]University of Belgrade, Serbia; [3]Beth Israel Deaconess Medical Center/Harvard Medical School, Boston, MA, USA; [4]Neurologia Śląska Centrum Medyczne, Katowice, Poland; [5]University Hospital, Krakow, Poland; [6]Dianthus Therapeutics, NY, USA; [7]Truman Statistical Services, Sydney, Australia; [8]Rambam Medical Center, Haifa, Israel; [9]University of Copenhagen, Copenhagen, Denmark.

MAIN FINDINGS

- Claseprubart treatment was well tolerated and resulted in clinically meaningful and statistically significant improvements across key assessments.
- The benefit/risk profile of both doses was similar, supporting research focus on the lower dose.
- Claseprubart has the potential to deliver meaningful benefits to AChR Ab+ gMG patients via infrequent self-administered SC injections and a reduced risk of encapsulated bacterial infections versus C5 inhibitors.
- Claseprubart 300mg Q2W and Q4W will be evaluated in an upcoming Phase 3 trial in gMG.

INTRODUCTION

- Generalized myasthenia gravis (gMG) is a chronic autoimmune disease caused by immunoglobulin G (IgG) autoantibodies targeting the acetylcholine receptor (AChR), leading to activation of the classical compliment pathway and damage to the neuromuscular junction.[1]
- Complement inhibition is an established mechanism for the treatment of gMG, and several downstream inhibitors of the complement cascade that target C5 are approved in the United States and Europe.[2–4]
- Though effective for many patients, these therapies either require intravenous (IV) infusions or daily injections. Additionally, by targeting C5, these therapies block all three complement pathways and potentially increase the risk of acquiring life-threatening encapsulated bacterial infections.[5]
- Claseprubart (DNTH103) is an investigational monoclonal antibody that specifically inhibits the classical complement pathway (CCP) upstream by selectively binding active C1s (aC1s).
- Unlike currently approved complement inhibitors, claseprubart offers a more focused treatment approach in gMG by only blocking the CCP and leaving the lectin and alternative pathways intact (**Figure 1**).
- Here, we report the results of the MaGic phase 2 study of claseprubart in patients with AChR antibody (Ab)+ gMG.

Figure 1. Claseprubart Mechanism of Action.

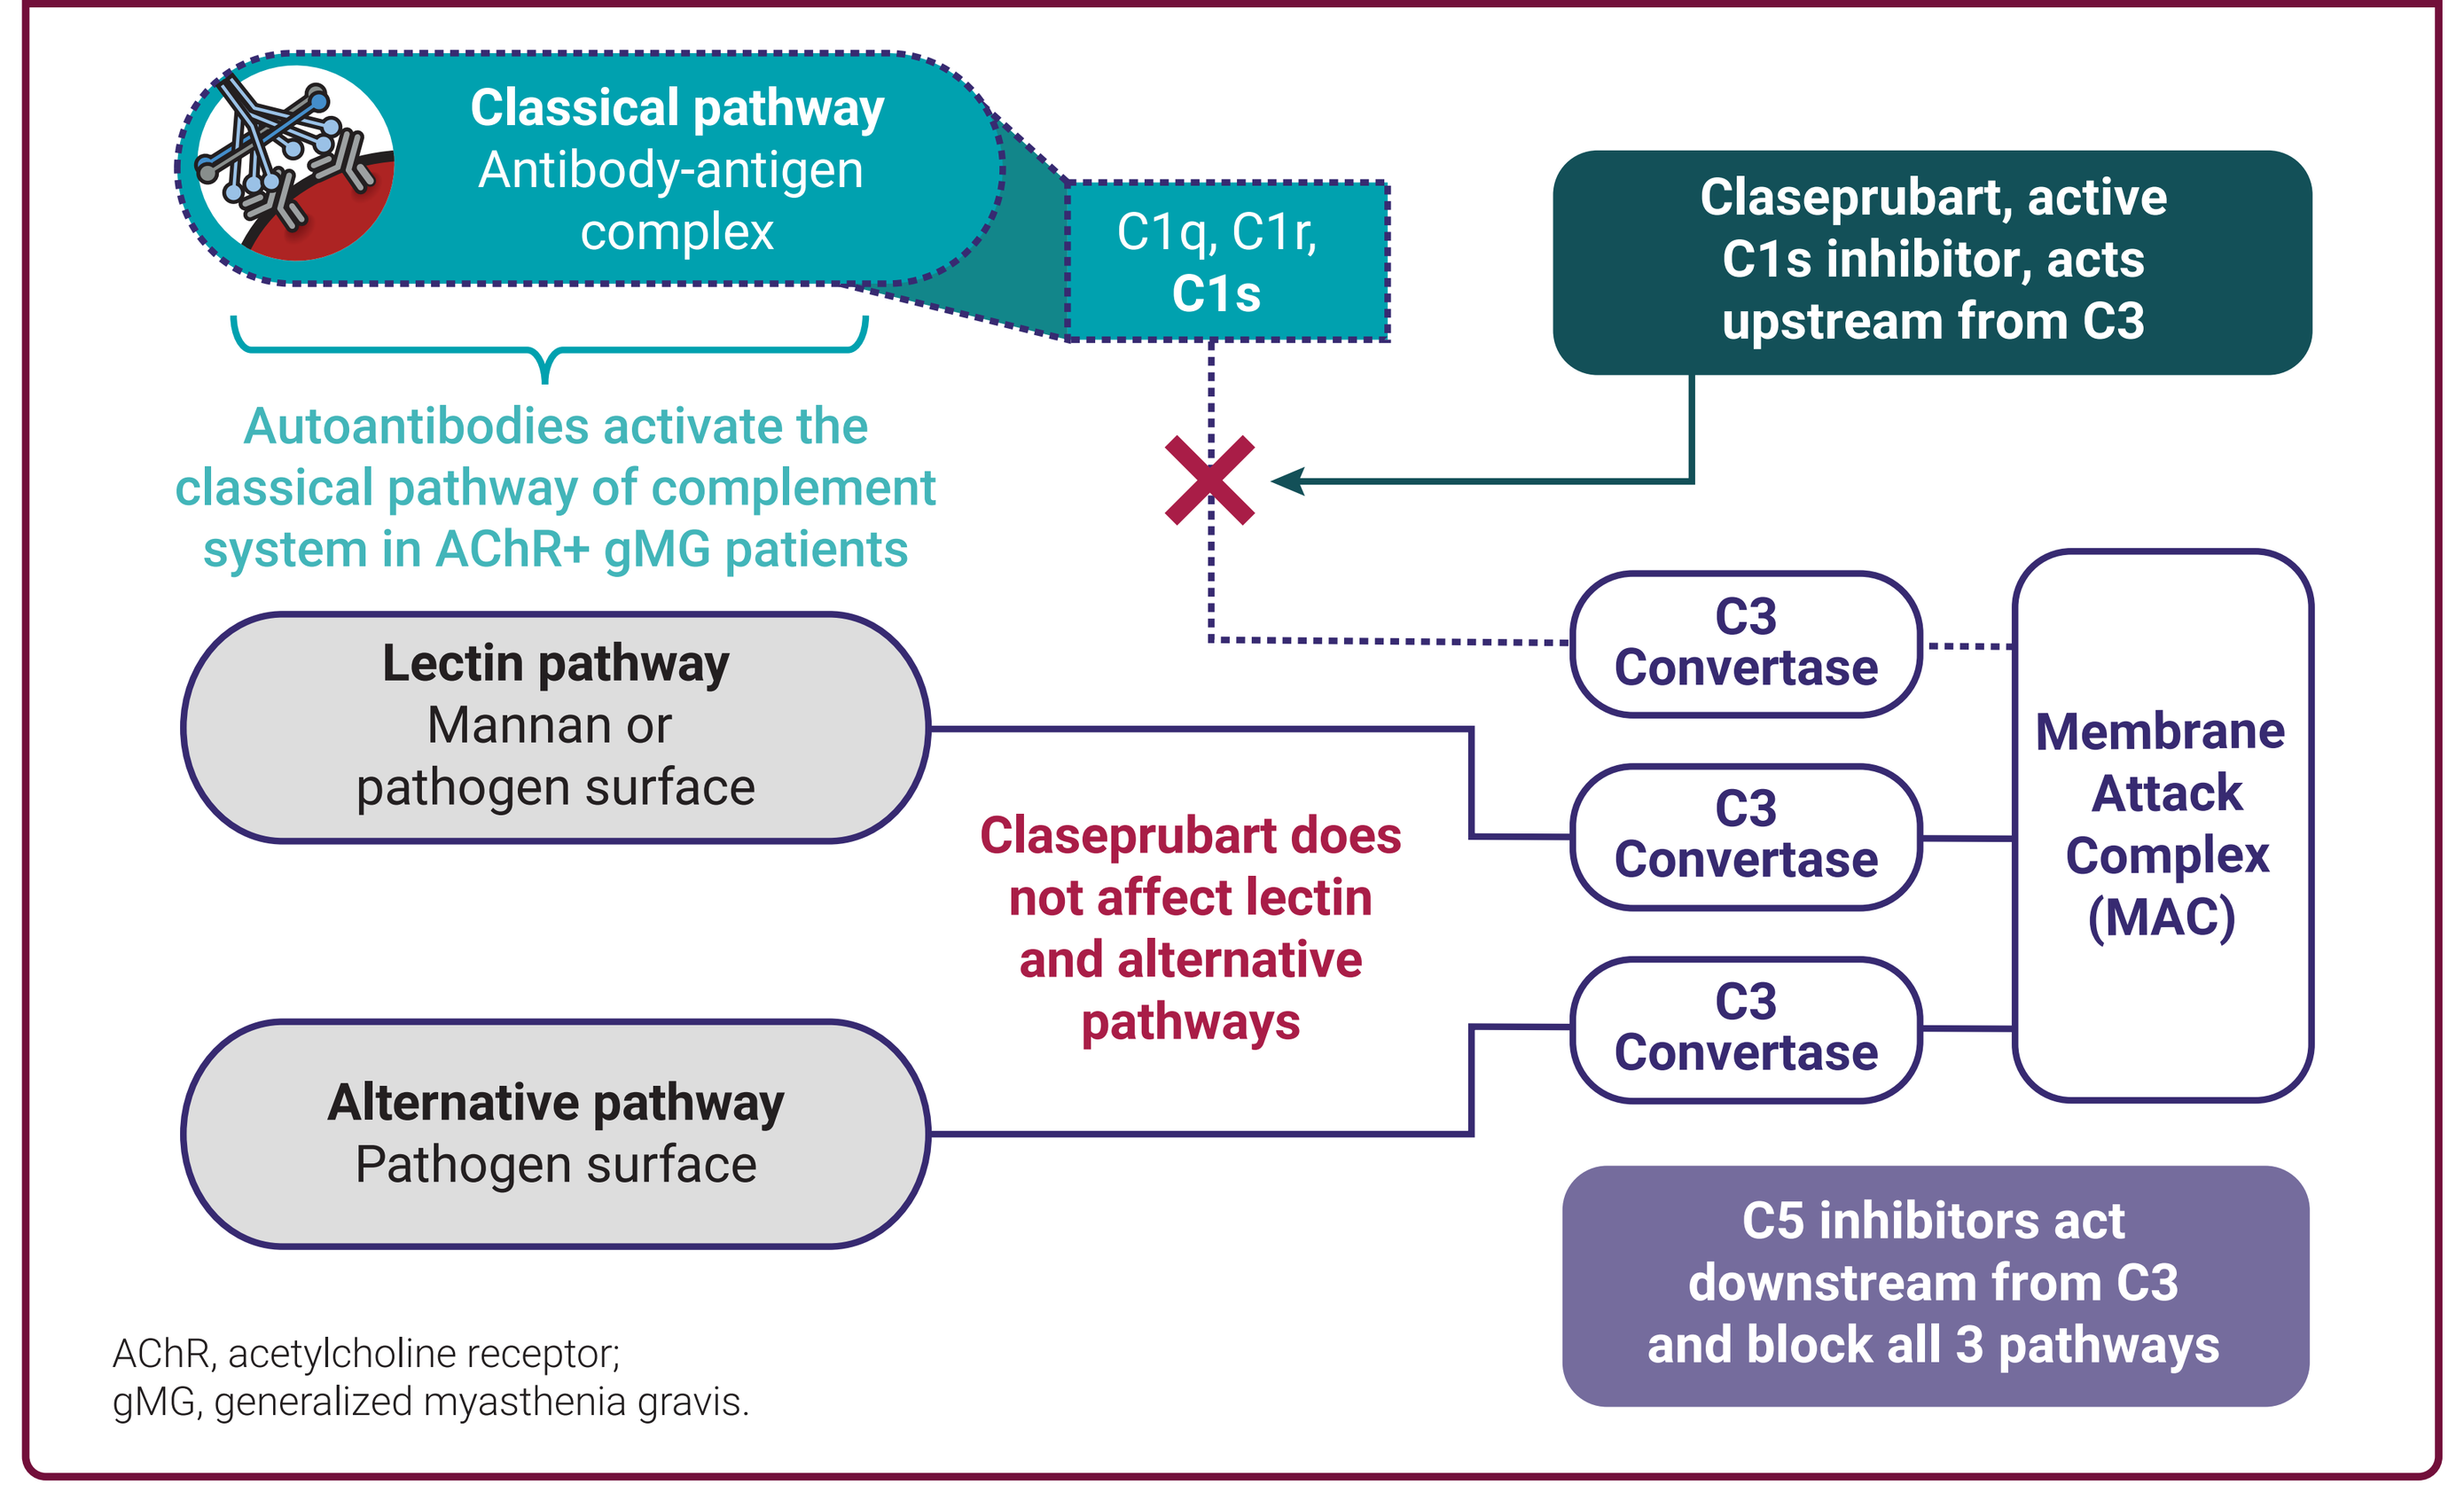
Classical pathway
Antibody-antigen complex
C1q, C1r, C1s
Claseprubart, active C1s inhibitor, acts upstream from C3
Autoantibodies activate the classical pathway of complement system in AChR+ gMG patients
Lectin pathway
Mannan or pathogen surface
Claseprubart does not affect lectin and alternative pathways
Alternative pathway
Pathogen surface
C3 Convertase
C3 Convertase
C3 Convertase
Membrane Attack Complex (MAC)
C5 inhibitors act downstream from C3 and block all 3 pathways

AChR, acetylcholine receptor; gMG, generalized myasthenia gravis.

OBJECTIVE

- To assess the safety and efficacy of subcutaneous claseprubart in patients with AChR Ab+ gMG.

METHODS

Study Design and Dosing

- MaGic is a global, multicenter, randomized, double-blind, placebo-controlled study to evaluate the safety, efficacy, pharmacokinetics, and pharmacodynamics of SC claseprubart (NCT06282159).
- Patients with AChR Ab+ gMG were enrolled and randomized 1:1:1 to receive claseprubart 300mg (Q2W), claseprubart 600mg (Q2W), or placebo for 13 weeks, followed by an ongoing 52-week open-label extension and 40-week safety follow-up (**Figure 2**).

Figure 2. MaGic Phase 2 Study Design

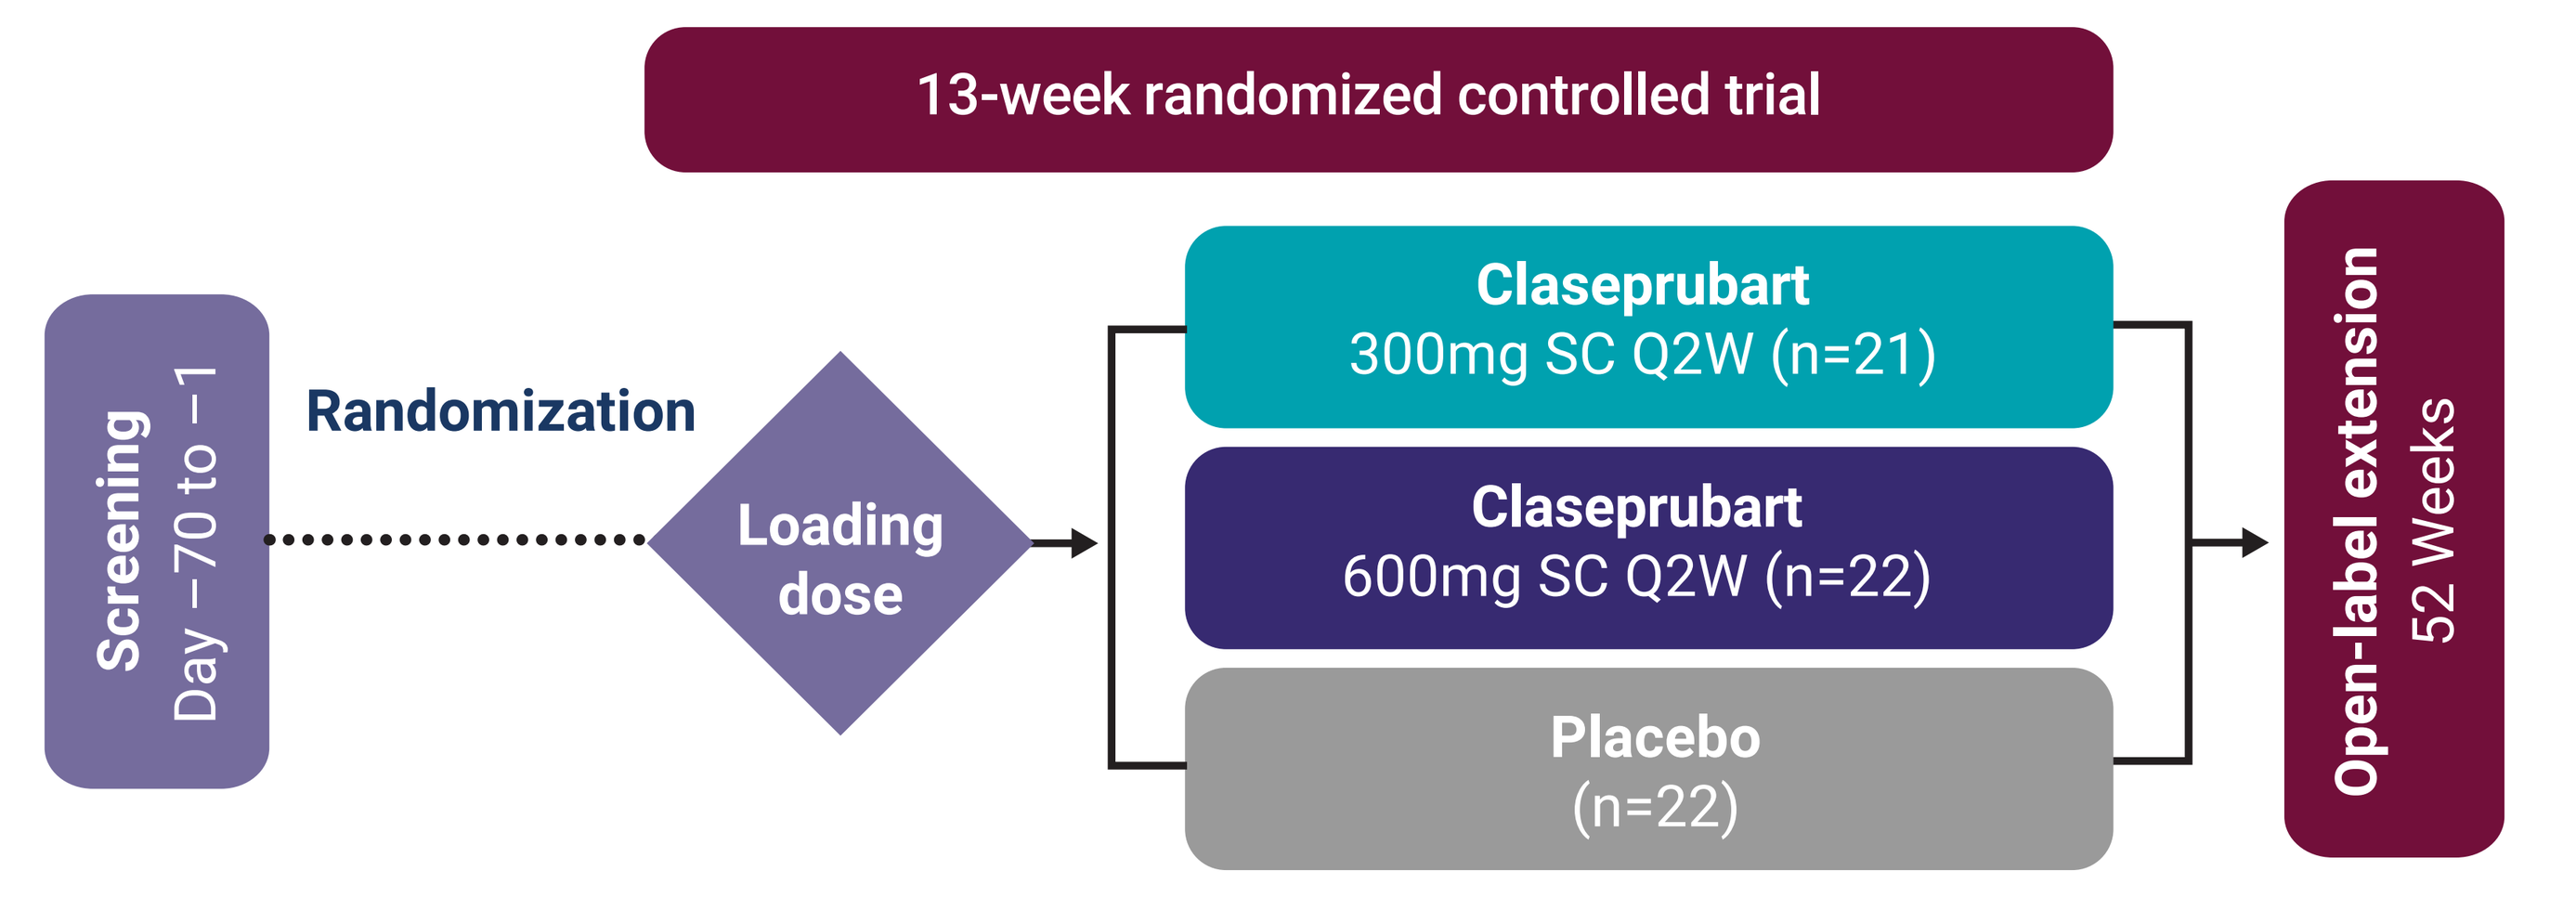
13-week randomized controlled trial
Screening
Day −70 to −1
Randomization
Loading dose
Claseprubart
300mg SC Q2W (n=21)
Claseprubart
600mg SC Q2W (n=22)
Placebo
(n=22)
Open-label extension
52 Weeks

Patients at least 18 years old with AChR antibody+ gMG were administered an IV loading dose of claseprubart followed by 300mg or 600mg SC Q2W. Study visits took place every subsequent 2 weeks with the final visit at Week 13 post-baseline. AChR, acetylcholine receptor; gMG, generalized myasthenia gravis; IV, intravenous; Q2W, once every 2 weeks; RCT, randomized controlled trial SC, subcutaneous.

Study Population

- Adults aged 18–75 years (inclusive) with a diagnosis of AChR Ab+ gMG, Myasthenia Gravis Foundation of American (MGFA) Class II-IVa at screening and randomization, and Myasthenia Gravis Activities of Daily Living (MG-ADL) score ≥6 at screening and randomization were eligible to participate in this study.
- Individuals with current or prior use of a complement inhibitor, use of an anti-FcRn agent within 90 days of randomization, use of rituximab within 6 months, or IVIg/PLEX within 4 weeks of randomization, or any known/untreated thymoma were not permitted to in the study.

Assessments

- The primary assessment for the study was to evaluate the safety and tolerability of claseprubart.
- Key secondary efficacy assessments included change from baseline in the MG-ADL Scale Score, the Quantitative Myasthenia Gravis (QMG) Scale score, and the Myasthenia Gravis Composite (MGC) Scale Score.
- Key exploratory assessments included achievement of minimal symptom expression (MSE; MG-ADL score ≤1), Myasthenia Gravis Composite Score (MGC), and Myasthenia Gravis Quality of Life 15-item scale (MG-QoL15r).

RESULTS

Disposition and Baseline Demographics

Table 1. Disposition and Baseline Demographics.

	Placebo (n=22)	Claseprubart 300mg Q2W(n=21)	Claseprubart 600mg Q2W (n=22)
Study discontinuations, n (%)	1 (4.5)	1 (4.8)	0 (0)
Due to AE, n (%)	0 (0)	0 (0)	0 (0)
Age, mean (SD), years	52.2 (16.5)	57.1 (13.7)	55.3 (12.0)
Male, n (%)	13 (59%)	14 (67%)	10 (45%)
Weight, mean (SD), pounds	195.0 (48.0)	192.5 (35.5)	179.0 (35.4)
Duration of disease, median (range), years	7.7 (0.4–21.2)	3.0 (0.5–22.1)	7.6 (1.0–37.3)
MG-ADL score at baseline, mean (SD)	8.5 (2.9)	8.2 (2.2)	8.4 (2.6)
QMG score at baseline, mean (SD)	14.2 (5.8)	12.2 (2.7)	12.2 (3.6)
MGFA class at screening, n (%)			
II	7 (32%)	11 (52%)	12 (55%)
III	12 (55%)	10 (48%)	9 (41%)
IVa	3 (14%)	0 (0%)	1 (5%)

AE, adverse event; MG-ADL, Myasthenia Gravis Activities of Daily Living scale; MGFA, Myasthenia Gravis Foundation of America clinical classification; Q2W, once every 2 weeks; QMG, Quantitative Myasthenia Gravis score; SD, standard deviation.

- Sixty-three of 65 patients completed the 13-week randomized controlled portion of the study. No patients discontinued due to adverse events (AEs) (**Table 1**).
- Baseline characteristics were balanced across treatment groups.

Safety Assessments

- Claseprubart was generally well tolerated with a safety profile similar to the placebo group (**Table 2**).
- All treatment-related AEs with both doses of claseprubart were mild or moderate in severity, and no patients receiving claseprubart developed a serious AE related to treatment.
- No significant safety findings were observed regarding infections or autoimmune activation with either claseprubart dose group.

Table 2. Safety and Tolerability.

	Placebo (n=22)	Claseprubart 300mg Q2W(n=21)	Claseprubart 600mg Q2W (n=22)
Any treatment-related AEs	13 (59.1)	15 (71.4)	21 (95.5)
Clinical AEs[a]	11 (50.0%)	13 (61.9%)	15 (68.2%)
Treatment-related serious AEs	1 (4.5%)	0 (0%)	0 (0%)
Infections	10 (45.5%)	5 (23.8%)	6 (27.3%)
Treatment-related serious infections	1 (4.5%)	0 (0%)	0 (0%)
Injection site reactions[b]	0 (0%)	2 (9.5%)	2 (9.1%)
Newly positive for ANA[c]	0 (0%)	1 (5.9%)	8 (36.4%)
Rashes	0 (0%)	0 (0%)	0 (0%)
Arthralgia	1 (4.5%)	1 (4.8%)	0 (0%)

[a]Excludes events in the investigations System Organ Class (MedDRA). [b]All injection site reactions were mild to moderate. [c]Represents participants who were ANA negative at baseline and tested positive at ≥1:320 at any point during RCT (percentages calculated from n=17 for 300mg arm and n=22 for 600mg arm). An ANA titer of ≥1:320 was an exclusion criterion for the clinical trial protocol. At end of RCT (Week 13), 2 of the 8 patients in 600mg arm tested negative for ANA, 2 of the 8 patients in 600mg arm remained positive but at <1:320. AE, adverse event; ANA, anti-nuclear antibodies; Q2W, once every 2 weeks; RCT, randomized control trial.

Efficacy Assessments

- At Week 13, both dose groups of claseprubart demonstrated rapid, sustained improvements in MG-ADL score from baseline compared to placebo (**Figure 3A**).
 - Significant improvement from baseline in MG-ADL was observed as early as Week 1 in 300mg (p=0.0906 vs. placebo) and 600mg (p=0.0211) groups compared to placebo (one-sided p-values, p<0.1 considered nominally significant).
 - Greater than 60% of patients achieved ≥5 point improvements in MG-ADL in both claseprubart dose groups at Week 13 (**Figure 3B**).
 - **MG-ADL improvement in the 300mg claseprubart arm (dose used in the upcoming Phase 3 study) was also assessed in patients with QMG score ≥10 at baseline. In this sub-group, the claseprubart 300mg arm (n=18) demonstrated greater improvement in MG-ADL score (−3.0 placebo-adjusted; p=0.0006) compared to the placebo arm (n=16) at Week 13.**
- Robust improvements from baseline in QMG score were observed at Week 13 in both claseprubart dose groups compared to placebo (**Figure 3C**). Claseprubart produced rapid improvement as early as Week 1 (300mg [p=0.0042 vs. placebo]; 600mg [p=0.0418]) compared to placebo.
 - At least 60% of patients achieved ≥5 point improvements in QMG in both claseprubart dose groups at Week 13 (**Figure 3D**).
- At Week 13, significantly more claseprubart-treated patients achieved MSE in the 300mg (36.8%; p=0.0550) compared to placebo (13.6%) (one-sided p-values, p<0.1 considered nominally significant). Twenty-seven percent of claseprubart 600mg patients achieved MSE at Week 13 though not significantly different from placebo.
- MGC scores were significantly improved from baseline to Week 13 in the 300mg (−4.1 placebo-adjusted [80% CI −6.0, −2.3]; p=0.0023 vs. placebo) and 600mg (−4.3 placebo-adjusted [80% CI −6.1, −2.5]; p=0.0016).
- Patient-reported quality of life (MG-QoL15r) was also significantly improved from baseline to Week 13 with 300mg claseprubart (−5.0 [SD 3.96]; p=0.0414) compared to placebo (−2.7 [SD 4.48]). The claseprubart 600mg group also showed improved MG-QoL15r scores at Week 13 (−4.2 [SD 3.85]; p=0.1122), which was not significantly different from placebo.

Figure 3. Improvement in MG-ADL and QMG.

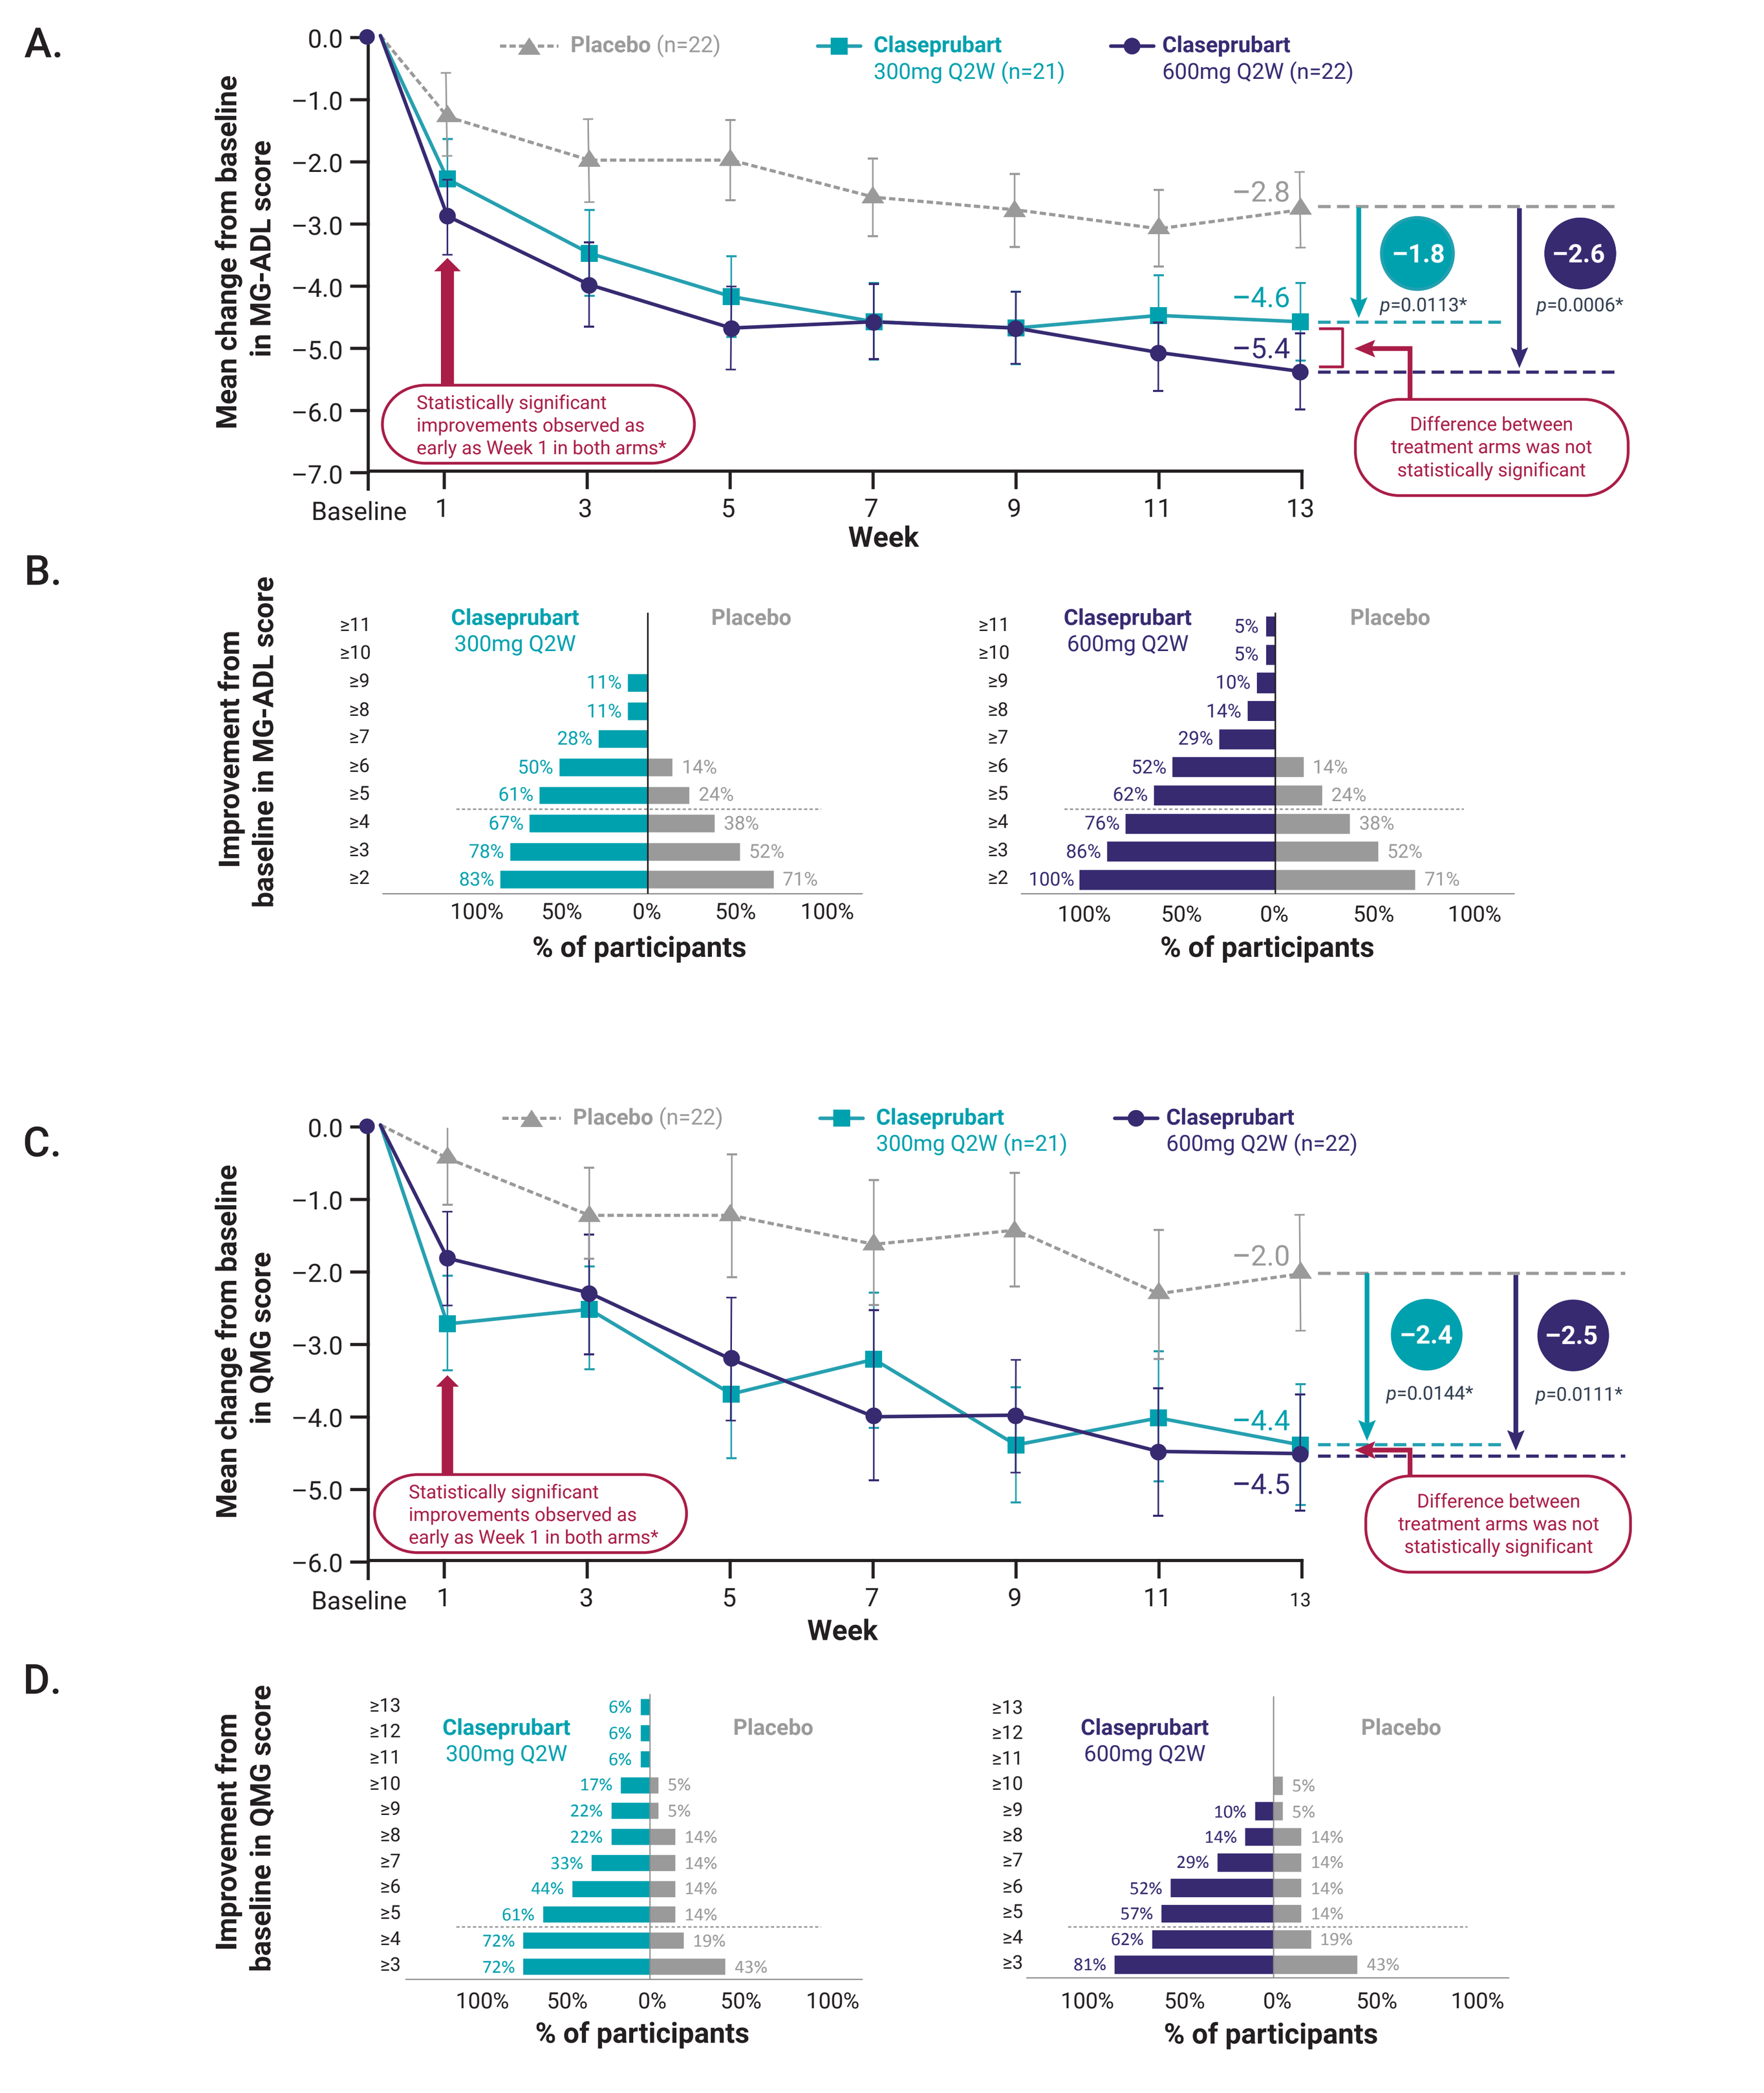
A.
Placebo (n=22)
Claseprubart 300mg Q2W (n=21)
Claseprubart 600mg Q2W (n=22)
Mean change from baseline in MG-ADL score
Week
−2.8
−4.6
−5.4
−1.8
p=0.0113*
−2.6
p=0.0006*
Statistically significant improvements observed as early as Week 1 in both arms*
Difference between treatment arms was not statistically significant
B.
Improvement from baseline in MG-ADL score
Claseprubart 300mg Q2W
Claseprubart 600mg Q2W
Placebo
% of participants
C.
Mean change from baseline in QMG score
−2.0
−4.4
−4.5
−2.4
p=0.0144*
−2.5
p=0.0111*
Statistically significant improvements observed as early as Week 1 in both arms*
Difference between treatment arms was not statistically significant
D.
Improvement from baseline in QMG score
% of participants

MG-ADL and QMG were assessed every other week, beginning 1 week after receiving study dose of claseprubart or placebo. (A, C) The change from baseline was analyzed using a mixed effect model for repeated measures (MMRM) with treatment group, visit, treatment by visit interaction, stratification factors, and baseline measure included. Bars represent standard error of the mean. (B, D) Magnitude of MG-ADL and QMG improvement was assessed at Week 13. *One-sided p-values are presented for comparisons of claseprubart vs placebo, with any p-value below 0.1 considered nominally statistically significant. MG-ADL, Myasthenia Gravis Activities of Daily Living scale; Q2W, once every 2 weeks; QMG, Quantitative Myasthenia Gravis score.

References

1. Michailidou I, et al. Front Immunol 2025;16:1526317
2. SOLIRIS (eculizumab) Prescribing Information
3. ULTOMIRIS (ravulizumab) Prescribing Information
4. ZILBRYSQ (zilucoplan) Prescribing Information
5. Halacova N, et al. Biologics 2026;6:3–21

Acknowledgements

The authors thank Aji Nair of Dianthus Therapeutics Inc. for assistance with the preparation of this poster, and Eastmond Medicomm Ltd for editing and production support, funded by Dianthus Therapeutics Inc.

Disclosures

SP, **PN**, **MS**, **AS**, **SS**, **TV** and **JV** have received grants, honoraria and/or consultancy fees from Dianthus. **MT** is a consultant for Dianthus. **CMB**, **US**, **SR**, and **LH** are employees of Dianthus. **SG** was an employee of Dianthus during the conduct of the trial.

Presented at the AAN Autoimmune Neurology Conference (ANC), 7–8 August, 2026, Houston, TX, USA

Dianthus Therapeutics Inc., New York, NY, USA