Minimal Symptom Expression with Claseprubart, an Active C1s Inhibitor, in Patients with Generalized Myasthenia Gravis

008

Said R. Beydoun,[1] Nils Erik Gilhus,[2] Amit Sachdev,[3] Maria Ait-Tihyaty,[4] Caitlin Briggs,[4] Uzma Siddiqui,[4] Luke Hickey,[4] Marianna Lalla,, Marek Śmilowski[5]

[1]University of Southern California, CA, USA; [2]Haukeland University Hospital, Bergen, Norway; [3]Michigan State University Health Care, East Lansing, MI, USA; [4]Dianthus Therapeutics, NY, USA; [5]Neurologia Śląska Centrum Medyczne, Katowice, Poland.

MAIN FINDINGS

- 300mg claseprubart-treated patients were four times more likely to achieve MG-ADL-MSE and six times more likely to achieve QMG-MSE versus placebo.
- Achievement of MSE was rapid and all 300mg claseprubart-treated patients who achieved MG-ADL-MSE maintained a remission-like state for at least 6 weeks.
- The results of this analysis further support the clinical efficacy of claseprubart in patients with AChR Ab+ gMG with the potential benefit of infrequent subcutaneous self-administration.

INTRODUCTION

- Generalized myasthenia gravis (gMG) is an autoimmune disease caused by autoantibodies targeting the acetylcholine receptor (AChR), leading to activation of the classical complement pathway (CCP) and damage to the neuromuscular junction.[1]
 - Patients with gMG experience a variety of physical symptoms related to muscle weakness throughout their lifetime, leading to lower quality of life compared to the general population.[2]
 - Achieving remission is an important goal and should be a key goal of any treatment for gMG.
- Claseprubart (DNTH103) is an investigational, clinical-stage, potent monoclonal antibody engineered to selectively target the CCP by inhibiting only the active form of the C1s protein, a clinically validated complement target (**Figure 1**). Claseprubart is enhanced with YTE half-life extension technology designed to enable a more convenient subcutaneous, infrequently dosed, self-administered injection.
 - Current complement inhibitor therapies may be burdensome for patients and health care providers due to frequent and inconvenient administration.[3–5]
- The Phase 2 MaGic study in patients with gMG showed significant improvements in efficacy outcomes with subcutaneous (SC) claseprubart compared to placebo while maintaining a favorable safety profile.[6]
 - Here, we evaluated whether claseprubart use resulted in reaching minimal symptom expression (MSE), a measure of disease remission in gMG.[7]

Figure 1. Claseprubart Mechanism of Action.

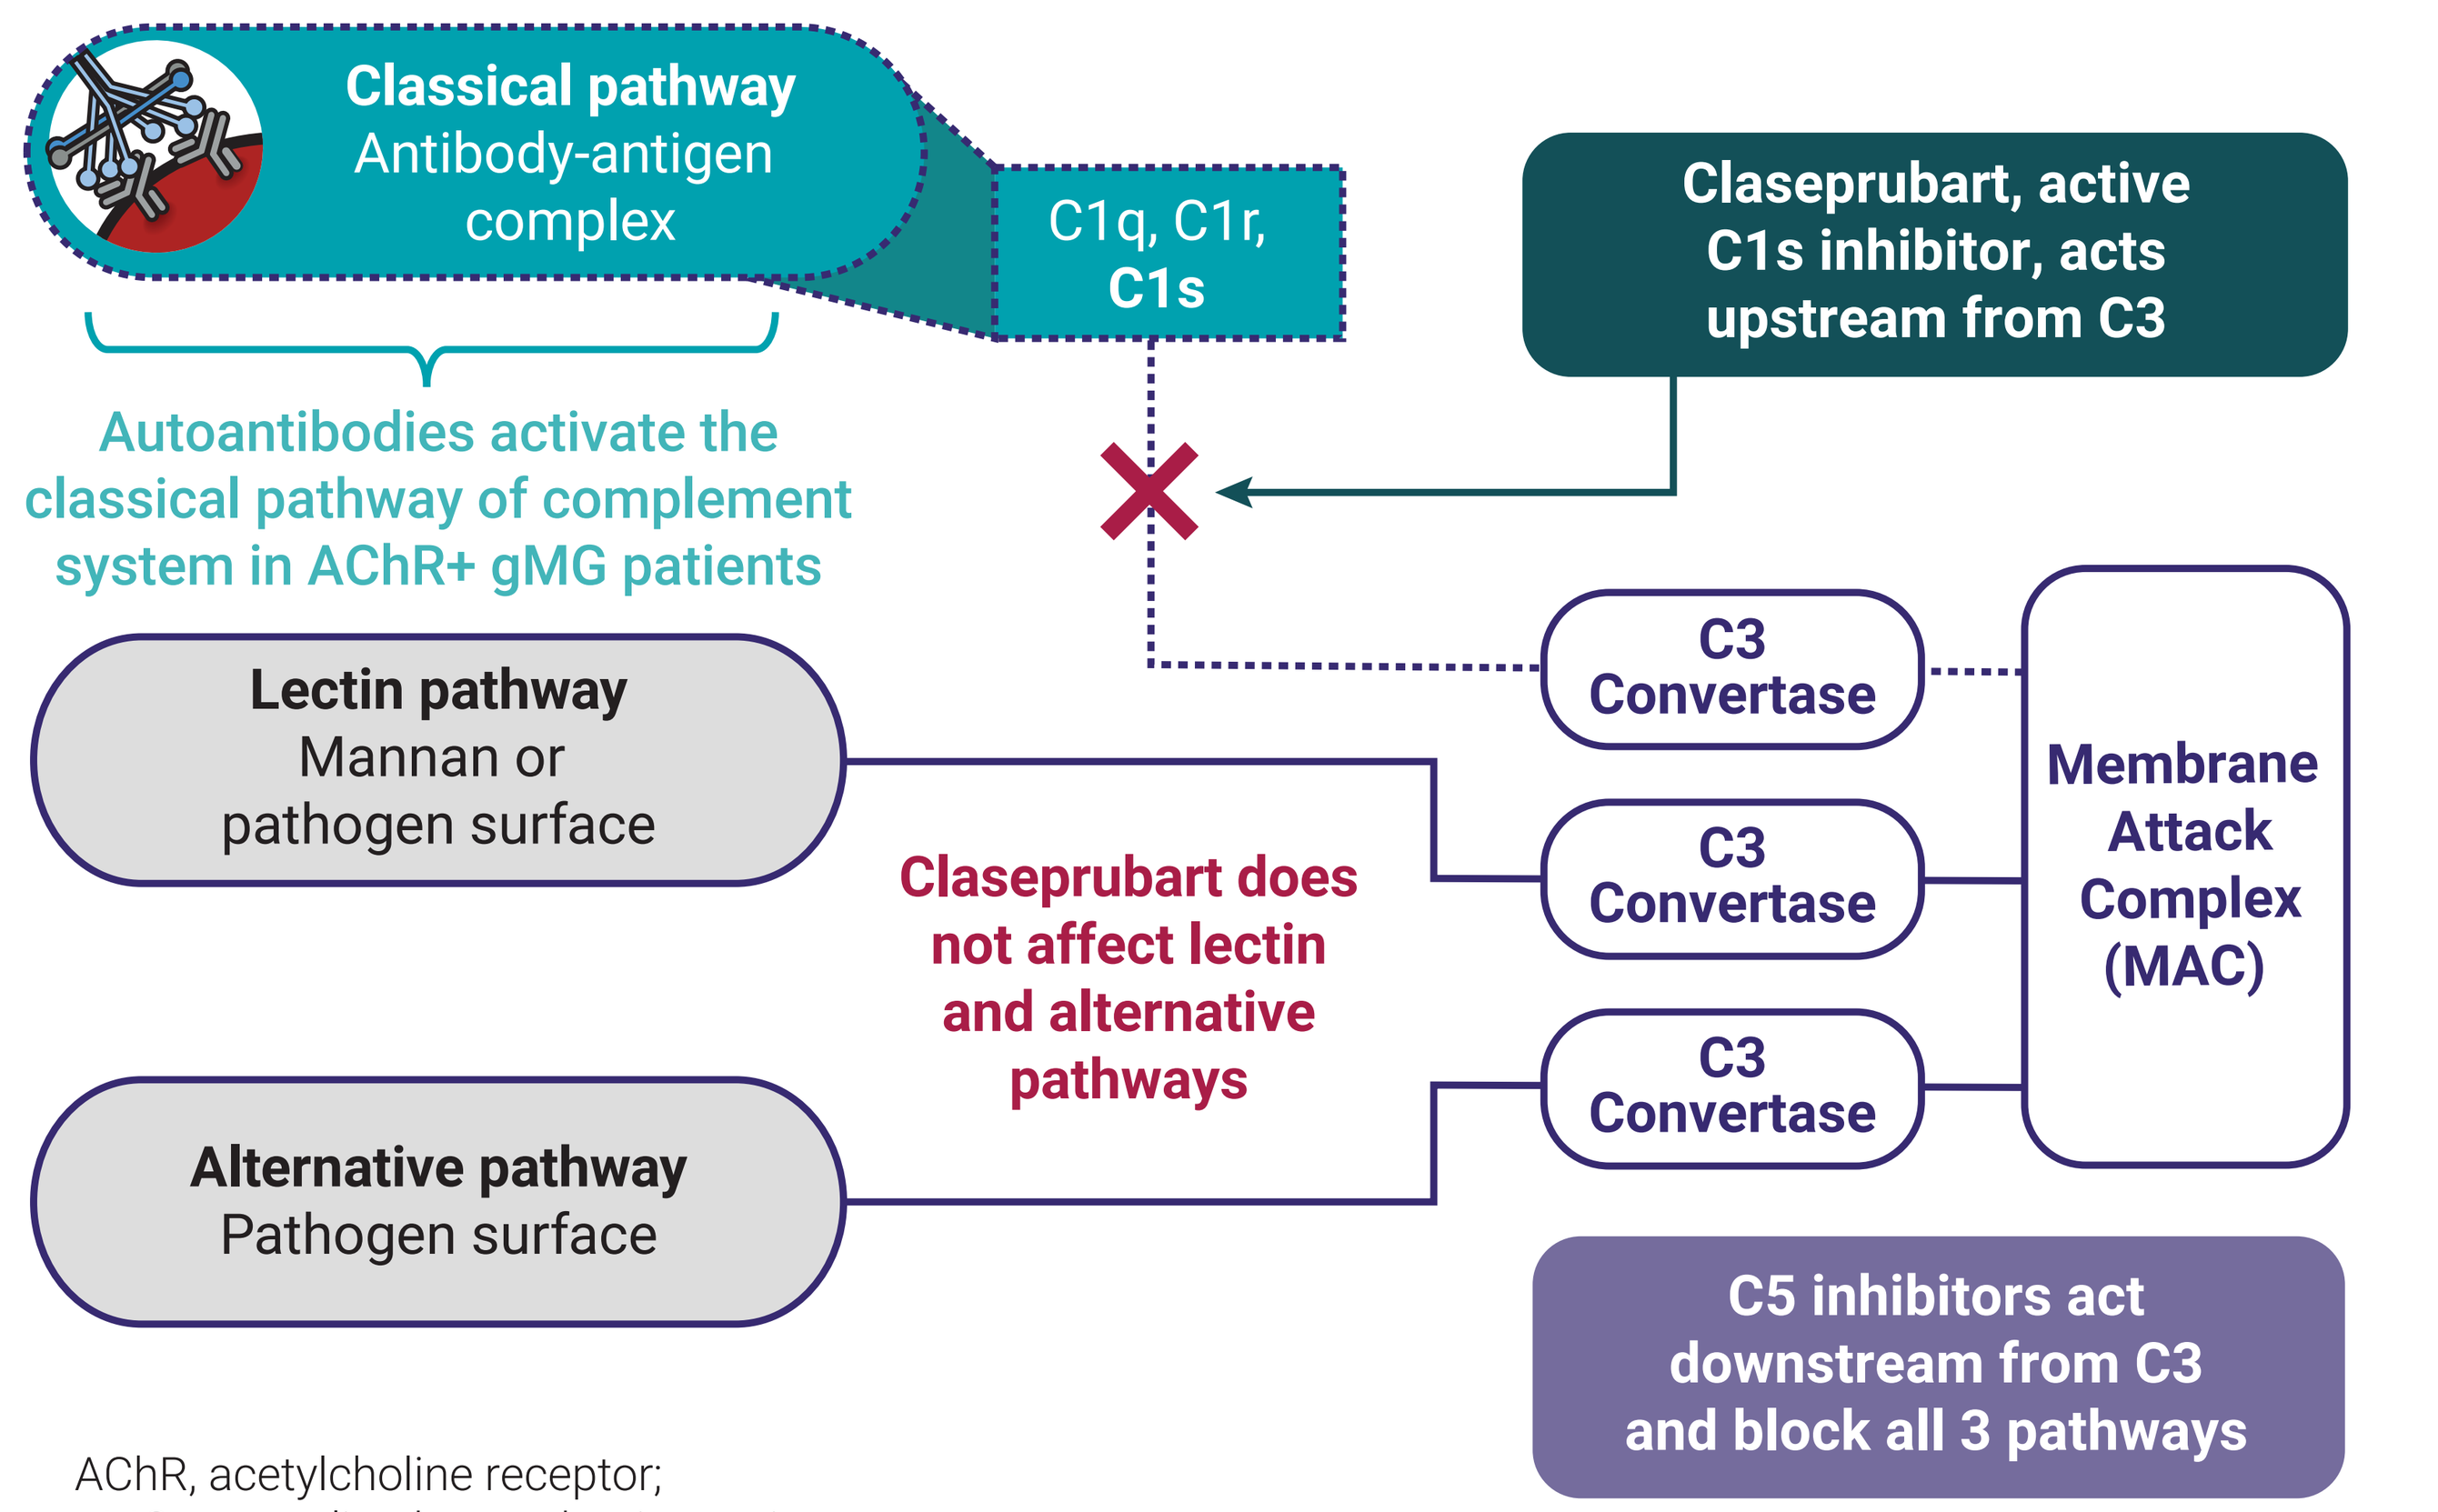
Classical pathway
Antibody-antigen complex
C1q, C1r, C1s
Autoantibodies activate the classical pathway of complement system in AChR+ gMG patients
Claseprubart, active C1s inhibitor, acts upstream from C3
Lectin pathway
Mannan or pathogen surface
Claseprubart does not affect lectin and alternative pathways
C3 Convertase
C3 Convertase
C3 Convertase
Membrane Attack Complex (MAC)
Alternative pathway
Pathogen surface
C5 inhibitors act downstream from C3 and block all 3 pathways

AChR, acetylcholine receptor; gMG, generalized myasthenia gravis.

OBJECTIVE

- To assess achievement of MSE in SC Q2W claseprubart-treated gMG patients in the MaGic study versus placebo.

METHODS

Study Design and Assessments

- MaGic is a global, multicenter, randomized, double-blind, placebo-controlled study to evaluate the safety, efficacy, pharmacokinetics, and pharmacodynamics of SC claseprubart (NCT06282159).
- Adults aged 18–75 years (inclusive) with a diagnosis of AChR Ab+ gMG were randomized 1:1:1 to receive claseprubart 300mg (Q2W), claseprubart 600mg (Q2W), or placebo for 13 weeks, followed by an ongoing 52-week open-label extension and 40-week safety follow-up (**Figure 2**).
- The primary objective for the study was to evaluate the safety and tolerability of claseprubart. Key secondary efficacy assessments included change from baseline in the Myasthenia Gravis Activities of Daily Living (MG-ADL) Scale Score and the Quantitative Myasthenia Gravis (QMG) Scale score.

Minimal Symptom Expression Analysis

- MG-ADL-MSE was defined as achieving an MG-ADL score of 0 or 1.
- QMG-MSE was defined as achieving a QMG score of ≤3.

Figure 2. MaGic Phase 2 Study Design.

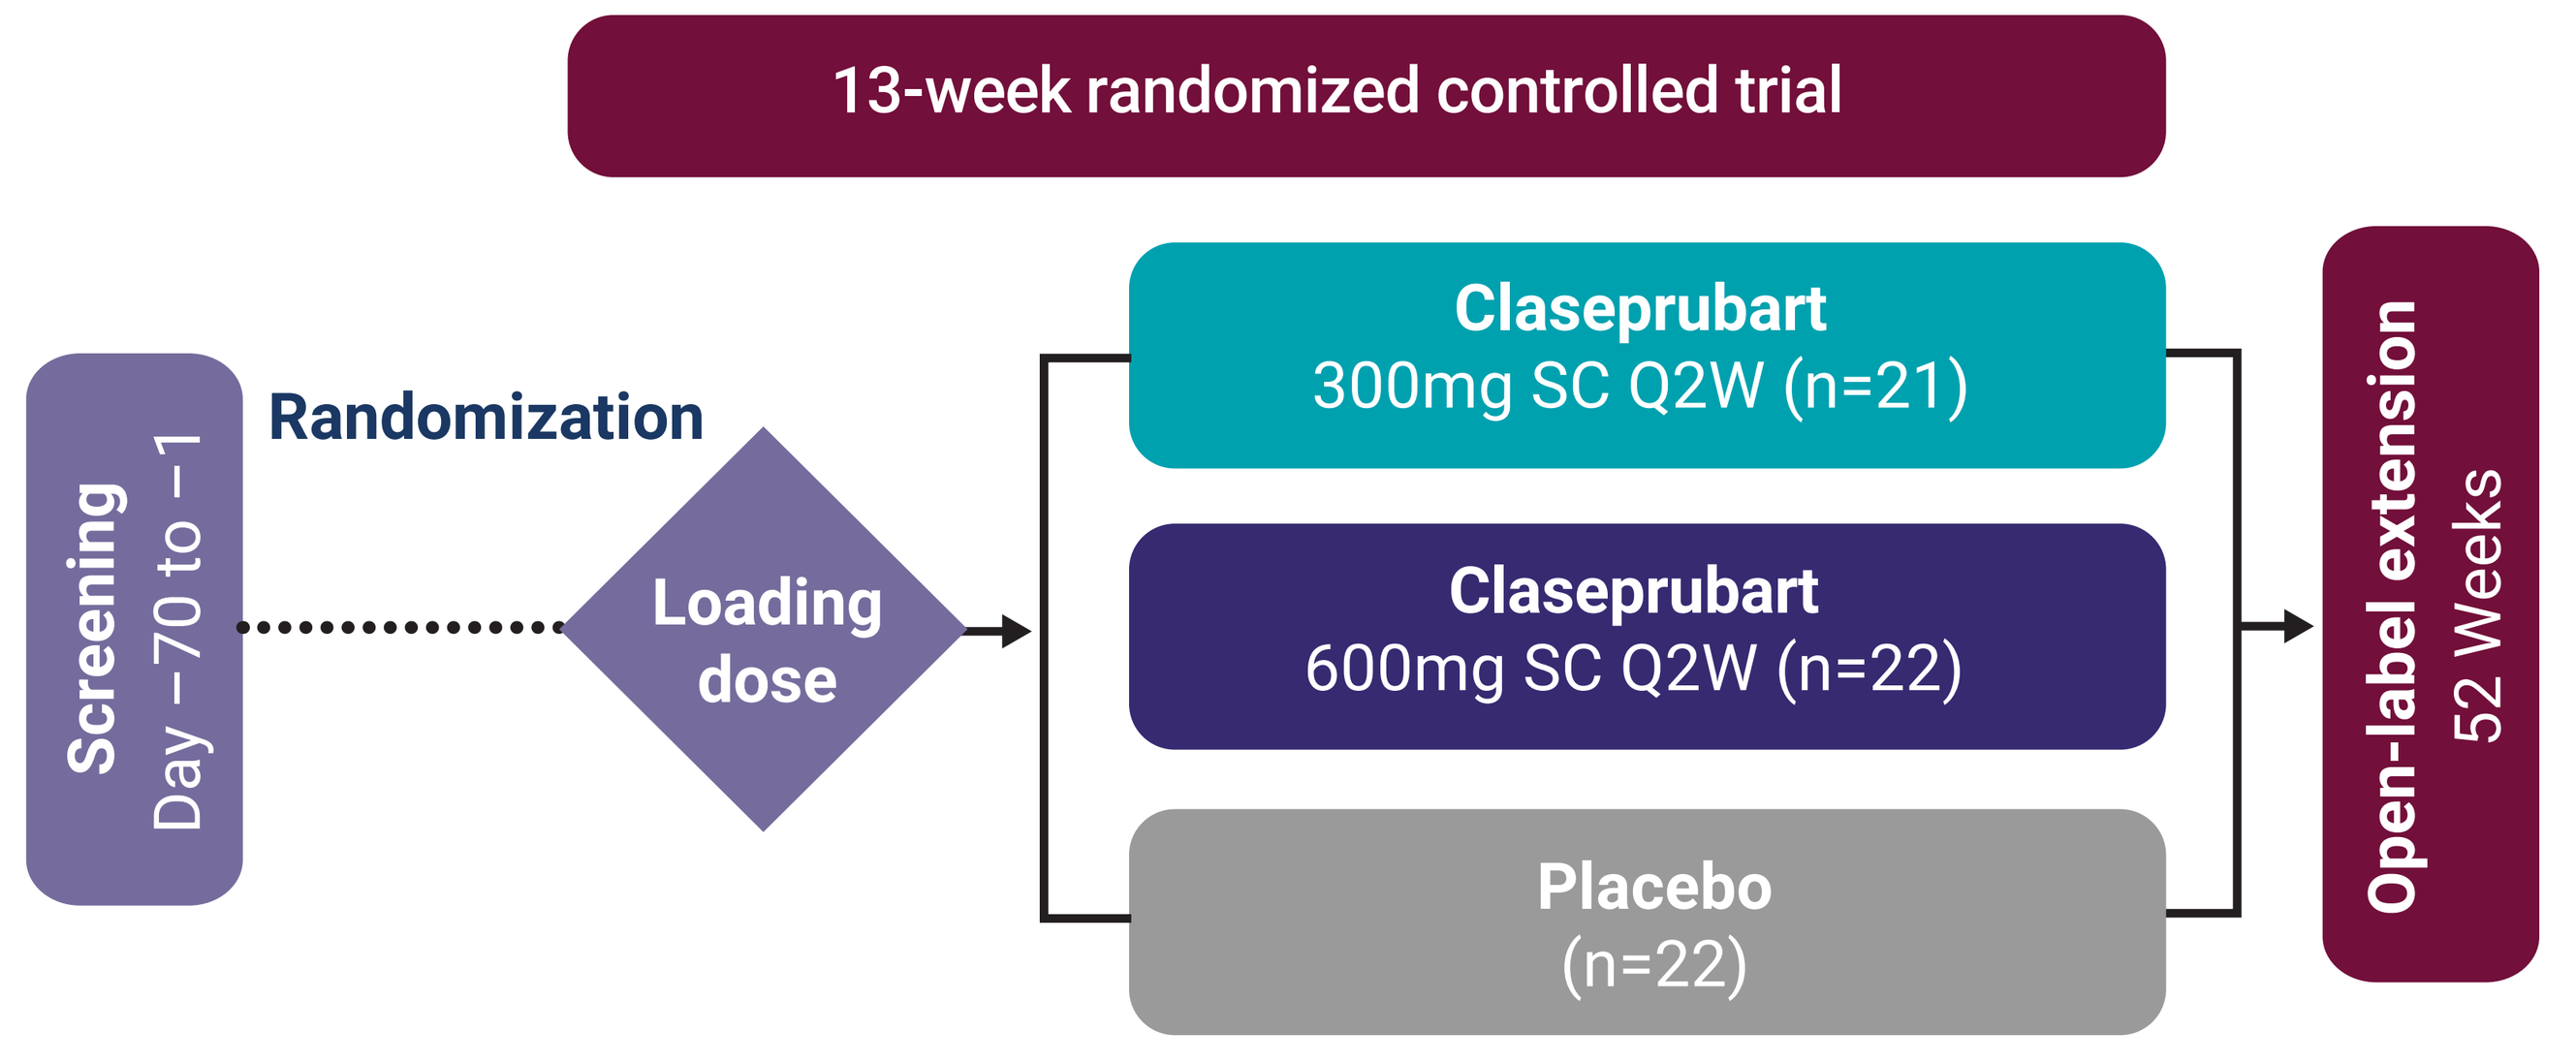
13-week randomized controlled trial
Screening
Day −70 to −1
Randomization
Loading dose
Claseprubart
300mg SC Q2W (n=21)
Claseprubart
600mg SC Q2W (n=22)
Placebo
(n=22)
Open-label extension
52 Weeks

Q2W, once every 2 weeks; SC, subcutaneous.

RESULTS

Baseline Characteristics and Disposition

- Sixty-five patients with AChR Ab+ gMG were randomized and received 300mg SC claseprubart (n=21), 600mg SC claseprubart (n=22), or placebo (n=22). Demographics across treatment arms were generally well-balanced. Approximately 57% of patients in the study were male with a mean age of 54.8 (standard deviation 14.1) years and a median disease duration of 5.6 years.
- Sixty-three patients completed the 13-week randomized controlled portion of the study. One placebo and one 300mg claseprubart patient discontinued, neither due to an adverse event.

Improvement in MG-ADL and QMG Scores

- MG-ADL score was significantly improved with both dose groups of claseprubart compared to placebo at Week 13 (**Figure 3A**).
 - Improvement was observed as early as Week 1 in 300mg (p=0.0908 vs. placebo) and 600mg (p=0.0211) groups compared to placebo (one-sided p-values, p<0.1 considered nominally significant).
 - Similarly, significant improvements in QMG score was observed in both claseprubart dose groups at Week 13 compared to placebo (**Figure 3B**).
 - Rapid improvement at Week 1 was observed with claseprubart (300mg [p=0.0042 vs. placebo]; 600mg [p=0.0418]) compared to placebo.

Figure 3. Change in MG-ADL and QMG score by Week.

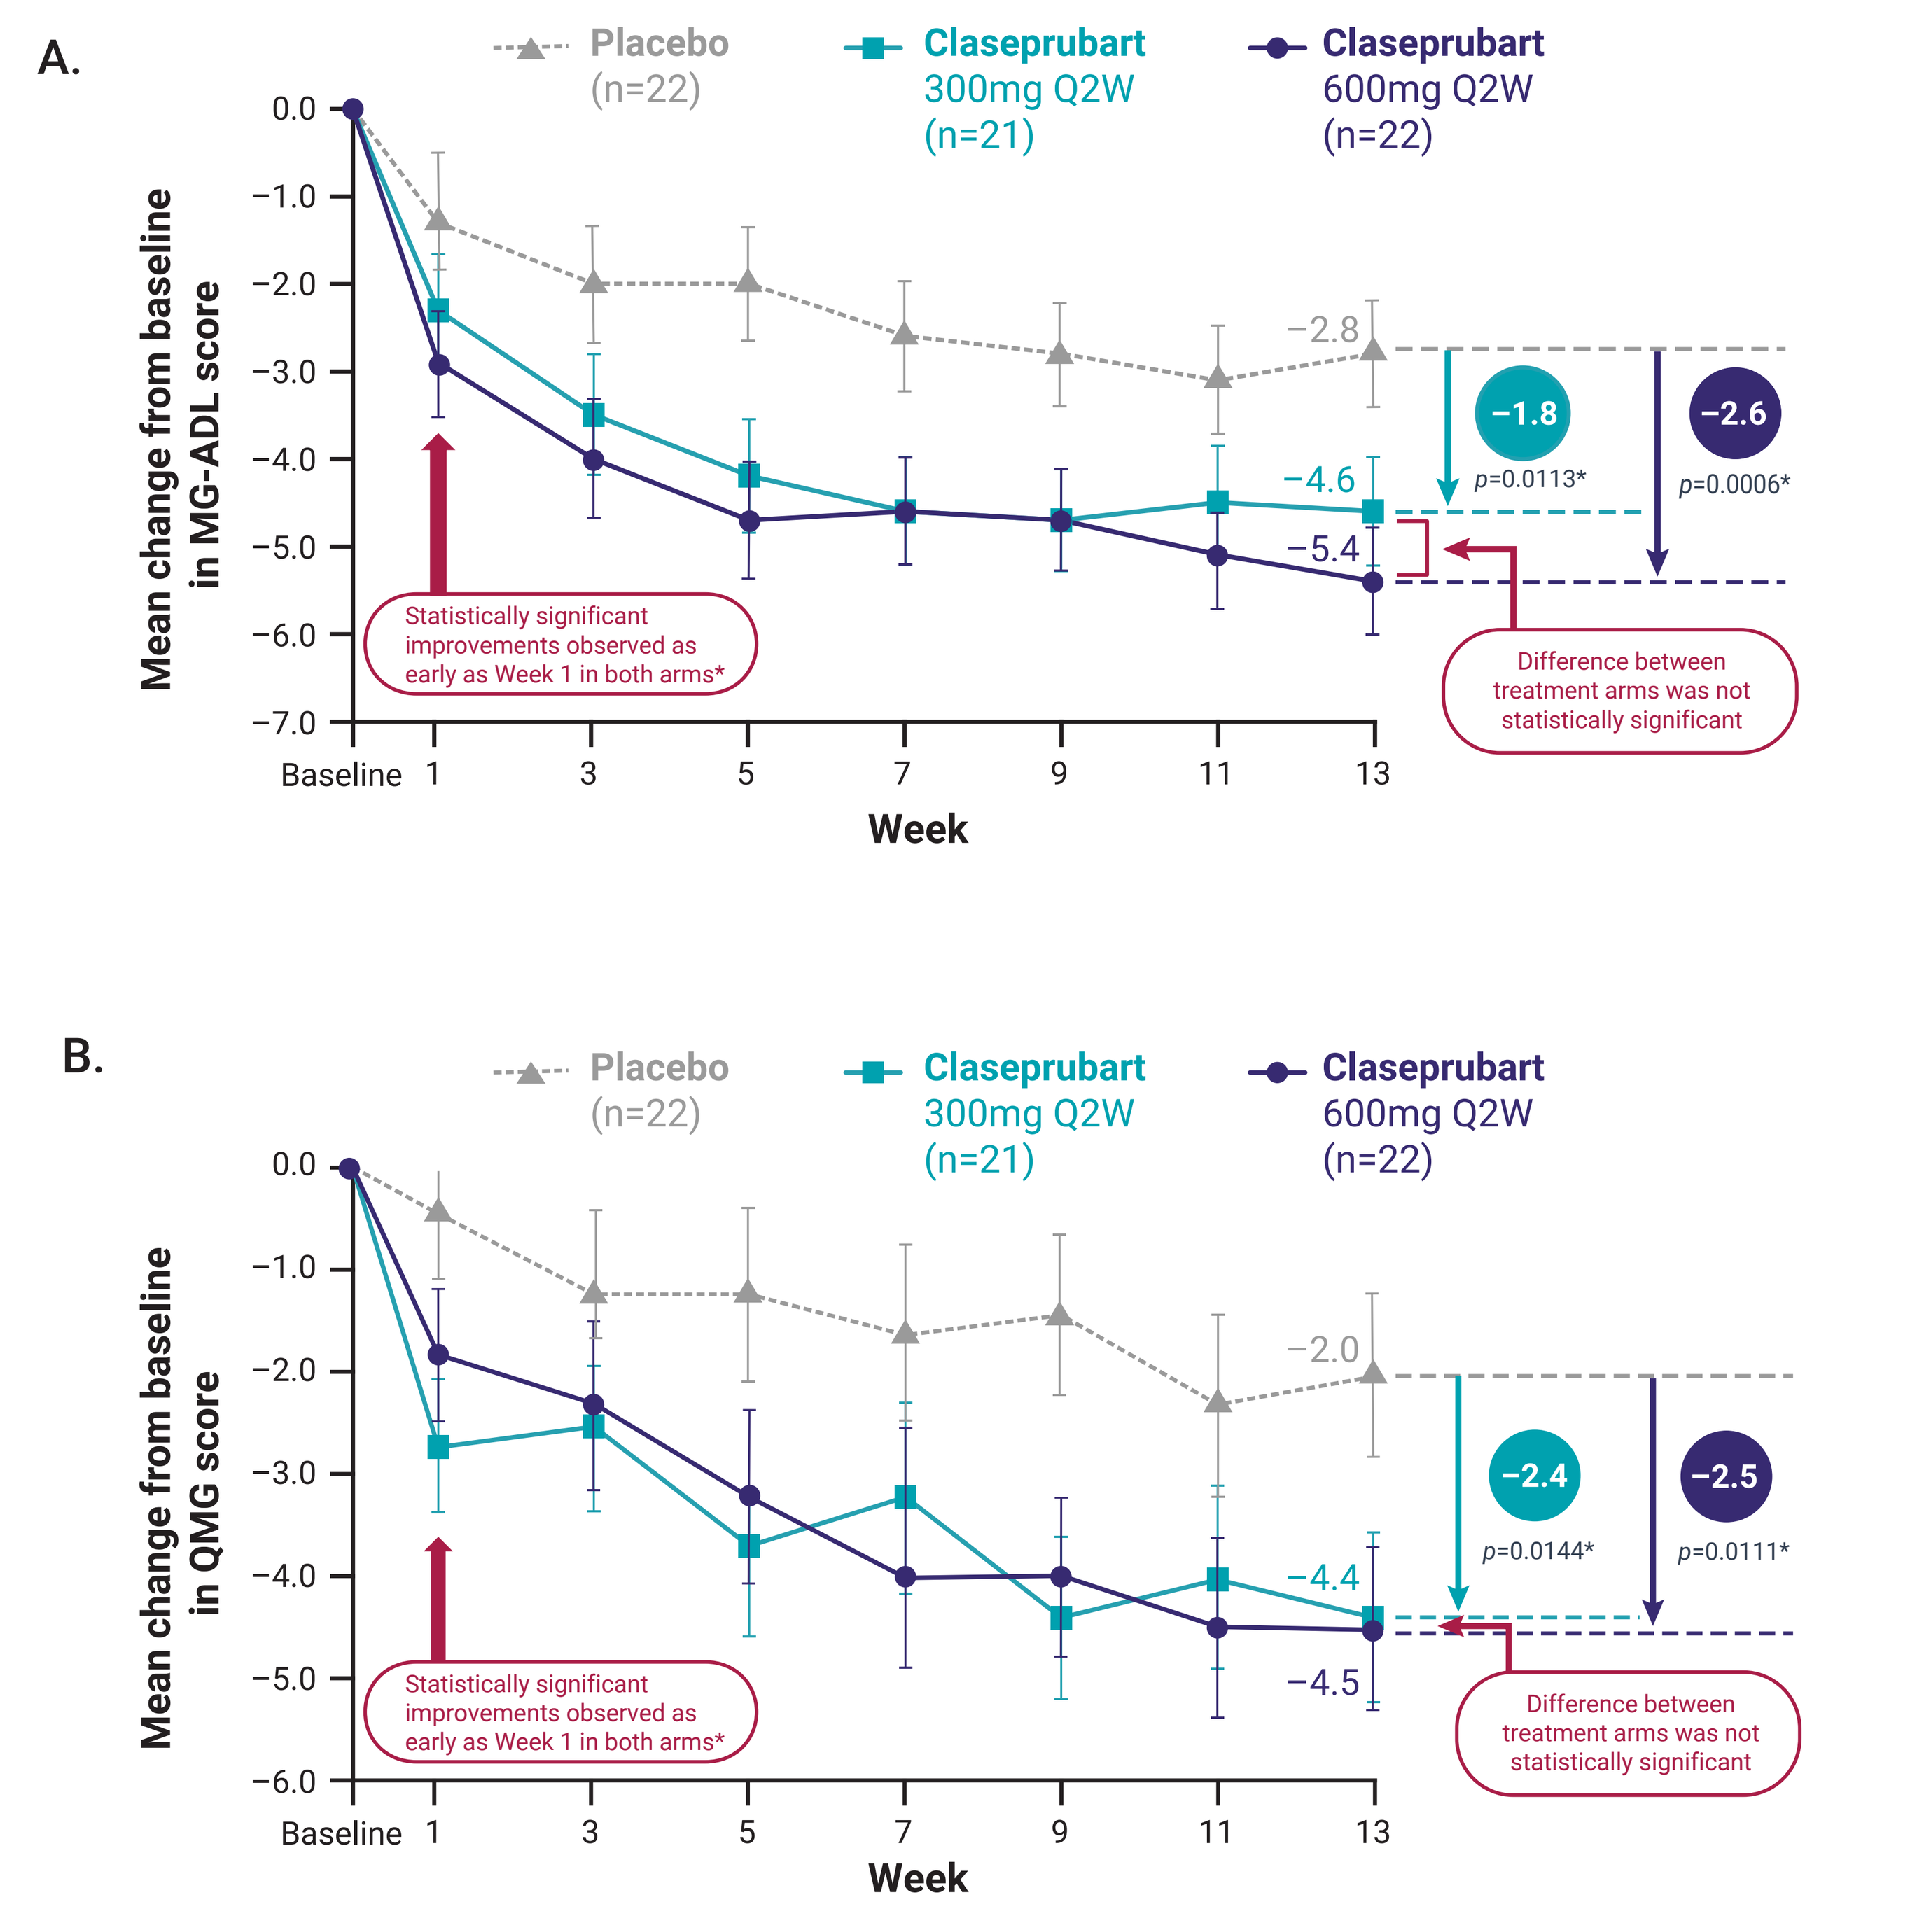
A.
Placebo (n=22)
Claseprubart 300mg Q2W (n=21)
Claseprubart 600mg Q2W (n=22)
Mean change from baseline in MG-ADL score
0.0
−1.0
−2.0
−3.0
−4.0
−5.0
−6.0
−7.0
Baseline
1
3
5
7
9
11
13
Week
−2.8
−4.6
−5.4
−1.8
p=0.0113*
−2.6
p=0.0006*
Statistically significant improvements observed as early as Week 1 in both arms*
Difference between treatment arms was not statistically significant
B.
Placebo (n=22)
Claseprubart 300mg Q2W (n=21)
Claseprubart 600mg Q2W (n=22)
Mean change from baseline in QMG score
0.0
−1.0
−2.0
−3.0
−4.0
−5.0
−6.0
Baseline
1
3
5
7
9
11
13
Week
−2.0
−4.4
−4.5
−2.4
p=0.0144*
−2.5
p=0.0111*
Statistically significant improvements observed as early as Week 1 in both arms*
Difference between treatment arms was not statistically significant

MG-ADL (A) and QMG (B) were assessed every other week, beginning 1 week after receiving study dose of claseprubart or placebo. The change from baseline was analyzed using a mixed effect model for repeated measures (MMRM) with treatment group, visit, treatment by visit interaction, stratification factors, and baseline measure included. Bars represent standard error of the mean. *One-sided p-values are presented for comparisons of claseprubart vs placebo, with any p-value below 0.1 considered nominally statistically significant.
MG-ADL, Myasthenia Gravis Activities of Daily Living scale; Q2W, once every 2 weeks; QMG, Quantitative Myasthenia Gravis score.

Minimal Symptom Expression

- At Week 13, significantly more patients from the 300mg claseprubart arm (37% [7/19], adjusted odds ratio (OR) 3.81, p=0.0550 vs. placebo) achieved MG-ADL-MSE compared to 14% (3/22) of placebo-treated patients (**Figure 4A**).
 - Achievement of MG-ADL-MSE was rapid with 300mg claseprubart, beginning at Week 5 (OR 4.60, p=0.0358 vs. placebo).
 - Median time to achieve MG-ADL-MSE was 3 weeks with 300mg claseprubart.
 - Of the 300mg claseprubart-treated patients who achieved MG-ADL-MSE at Week 13, all demonstrated MSE for at least 6 consecutive weeks.
- QMG-MSE was achieved by significantly more 300mg claseprubart-treated patients at Week 13 (32% [6/19], OR 5.84, p=0.0387 vs. placebo) compared to placebo-treated patients (9% [2/22]) (**Figure 4B**).
 - Median time to achieve QMG-MSE was 3 weeks with 300mg claseprubart.
 - Of the 300mg claseprubart-treated patients who achieved QMG-MSE at Week 13, five of six patients demonstrated MSE for at least 6 consecutive weeks.

Figure 4. Minimal Symptom Expression.

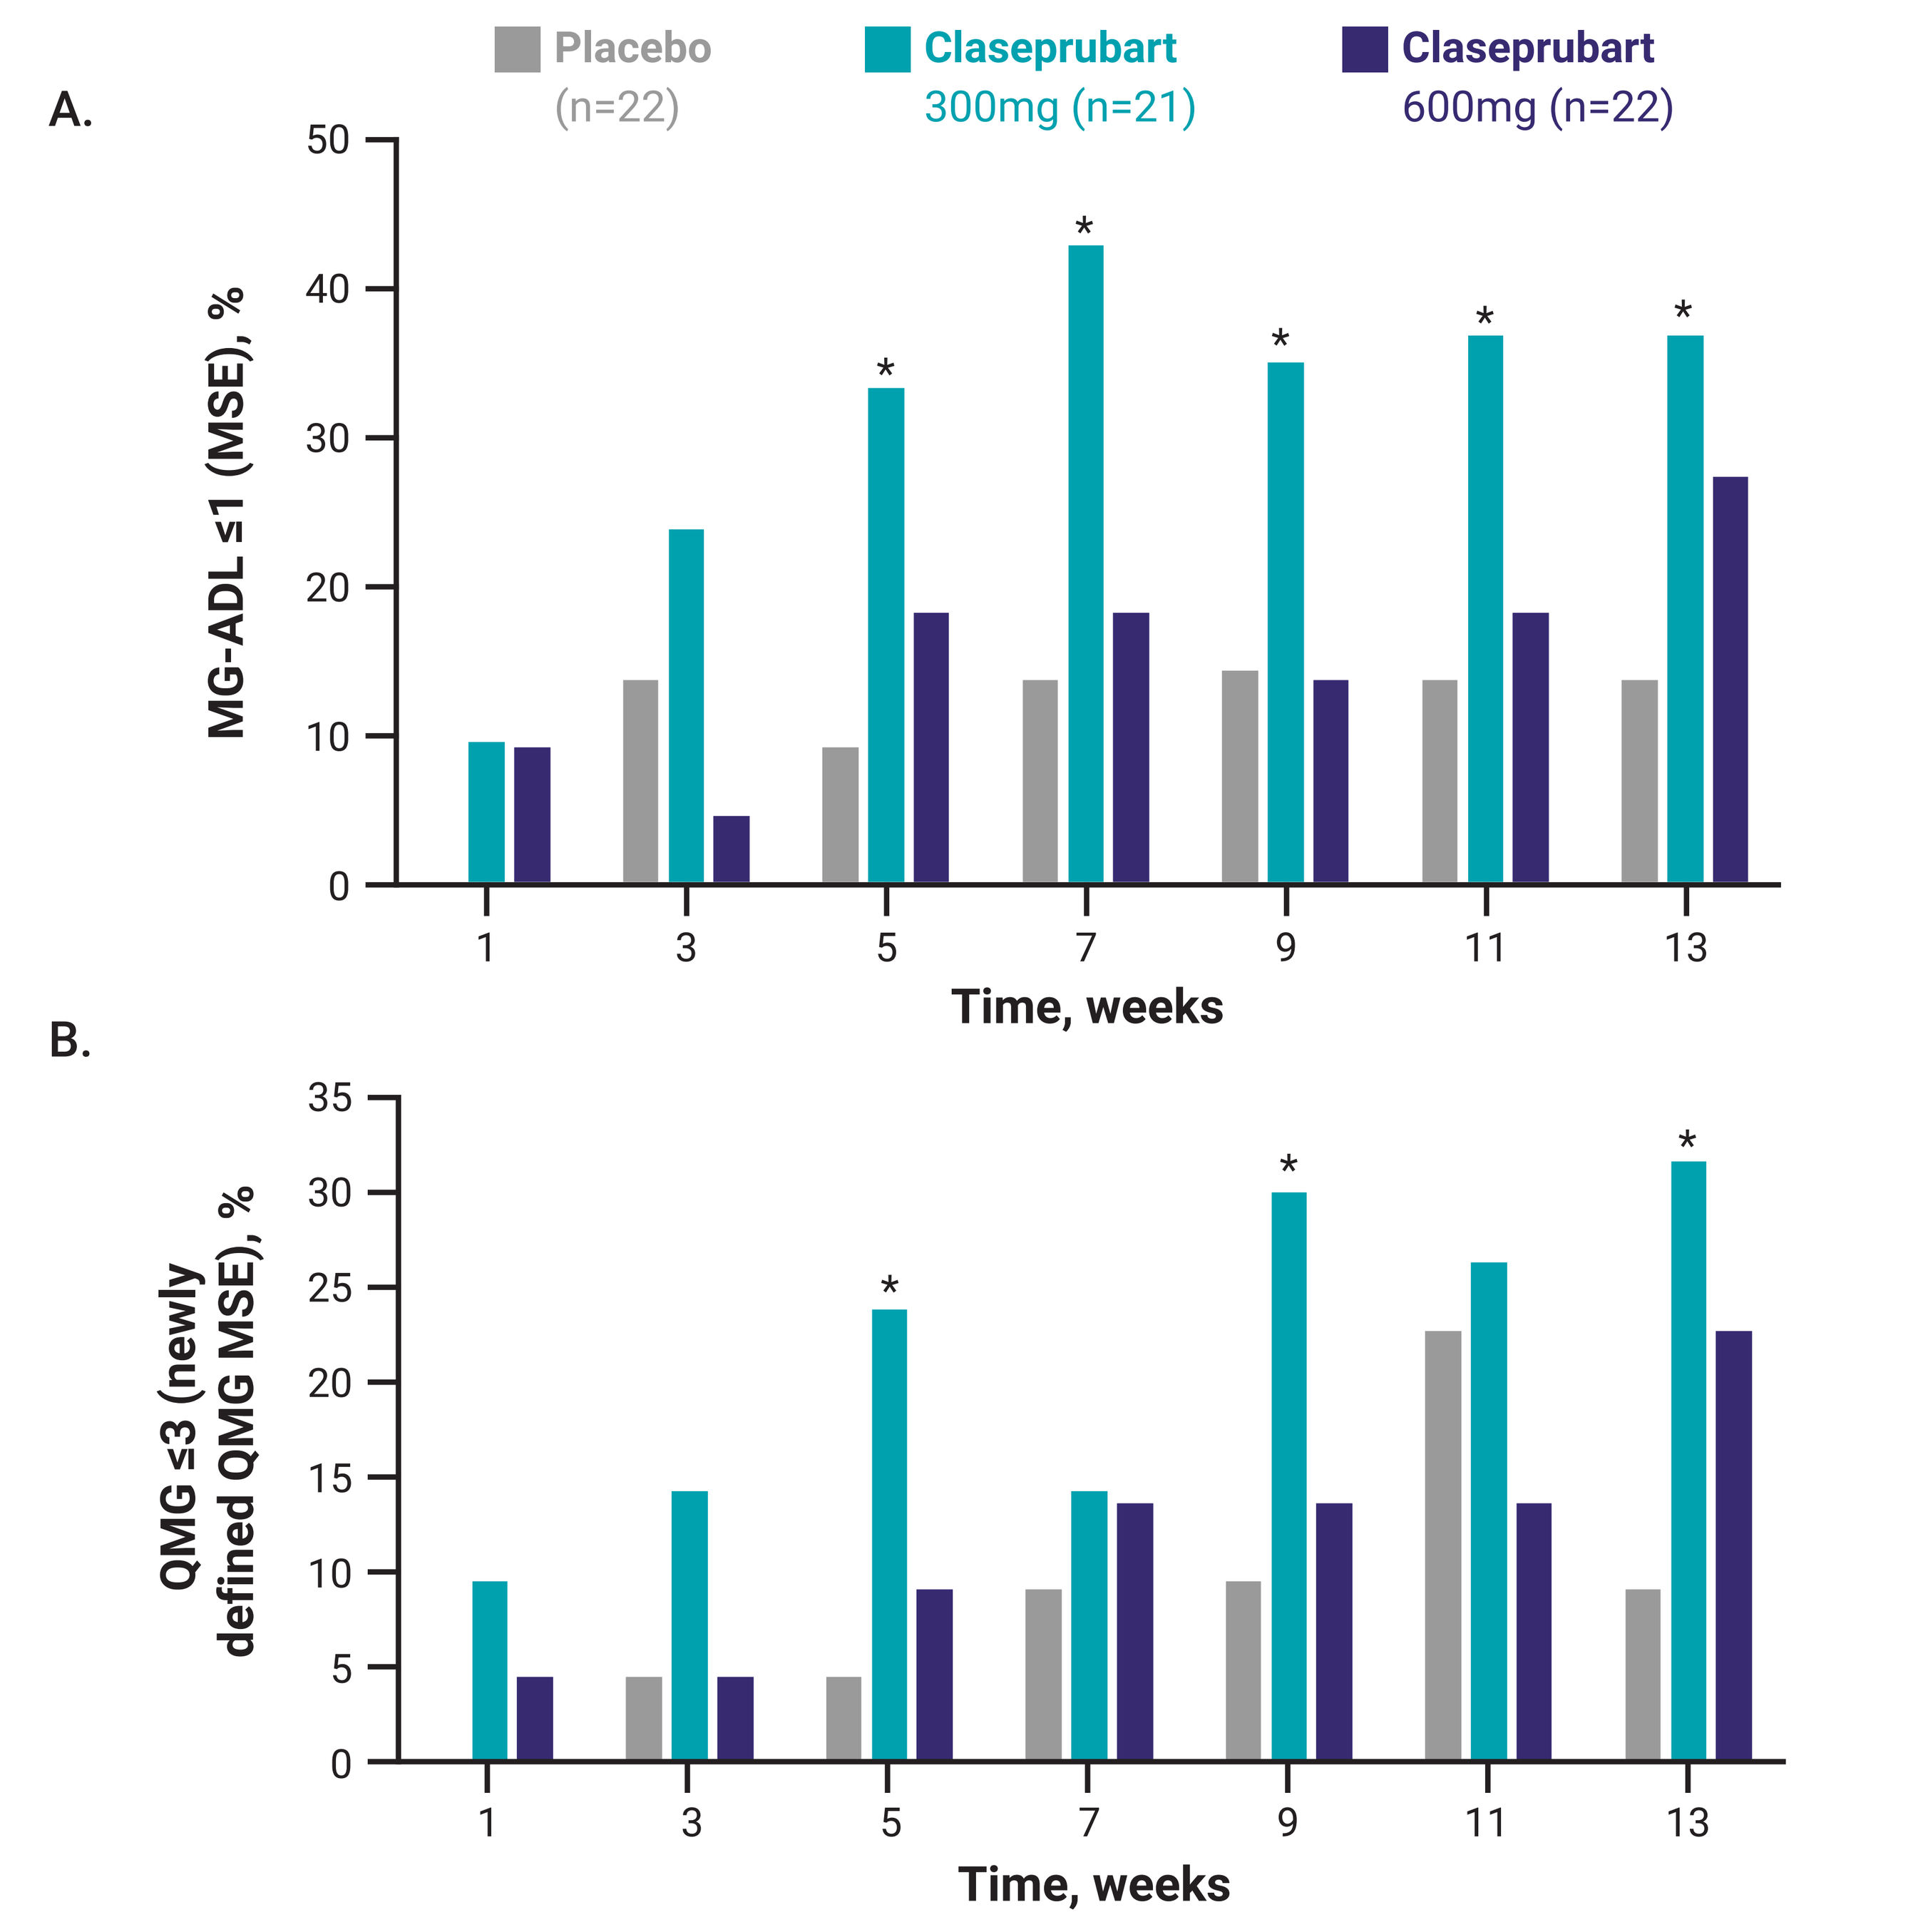
Placebo (n=22)
Claseprubart 300mg (n=21)
Claseprubart 600mg (n=22)
A.
MG-ADL ≤1 (MSE), %
50
40
30
20
10
0
1
3
5
7
9
11
13
Time, weeks
B.
QMG ≤3 (newly defined QMG MSE), %
35
30
25
20
15
10
5
0
1
3
5
7
9
11
13
Time, weeks

(A) MG-ADL-MSE was defined as achieving an MG-ADL score of ≤1. (B) QMG-MSE was defined as achieving a QMG score of ≤3. *One-sided p-values are presented for comparisons of claseprubart vs placebo, with any p-value below 0.1 considered nominally statistically significant. Adjusted analysis uses a logistic regression model with terms for treatment group, baseline and stratification factors. MSE, minimal symptom expression; MG-ADL, Myasthenia Gravis Activities of Daily Living scale; Q2W, once every 2 weeks; QMG, Quantitative Myasthenia Gravis score.

References

1. Howard J, et al. Ann New York Acad Sci 2017;1412;113–128
2. Chou M, et al. Muscle &. Nerve 2025;72:1035–1195
3. SOLIRIS (eculizumab) Prescribing Information
4. ULTOMIRIS (ravulizumab) Prescribing Information
5. ZILBRYSQ (zilucoplan) Prescribing Information
6. Narayanaswami P. AANEM 2025
7. Zhao R, et al. J Translat Med 2021;19:285

Acknowledgements

The authors thank Aji Nair of Dianthus Therapeutics Inc. for assistance with the preparation of this poster, and Eastmond Medicomm Ltd for editing and production support, funded by Dianthus Therapeutics Inc.

Disclosures

MS, **ENG** and **SRB** have received grants, honoraria and/or consultancy fees from Dianthus. **MAT**, **CMB**, **US**, **LH** and **ML** are employees of Dianthus.

Presented at the AAN Autoimmune Neurology Conference (ANC), 7–8 August, 2026, Houston, TX, USA

Dianthus Therapeutics Inc., New York, NY, USA